Abstract#: A17

The Dual Angiogenesis/Heparanase Inhibitor PG545 Inhibits Solid Tumor Progression in Xenograft Models of Breast, Prostate and Liver Cancer – a Comparative Assessment of Once- Versus Twice-Weekly Administration Schedules

Authors: [1]Edward Hammond, [1]Ian Bytheway, [1]Paul Handley, [2]Michelle Kappler, [2]Crystal McGirr, [2]Thomas J Gonda, [3]Maree Smith, [1]Keith Dredge. [1]Progen Pharmaceuticals Ltd, Brisbane, Australia; [2]Molecular Oncogenesis Group, Diamantina Institute for Cancer, Immunology and Metabolic Medicine, University of Queensland, Brisbane Australia; [3]Centre for Integrated Preclinical Drug Development, St Lucia Campus, University of Queensland, Brisbane, Australia.

PROGEN PHARMACEUTICALS

Web – www.progen-pharma.com
Contact – keithd@progen-pharma.com
(Keith Dredge, Director of Preclinical Development)
Australia PO Box 2403 Toowong Queensland 4066 Australia
T + 61 7 3842 3333

Introduction

PG545 is a fully sulfated synthetic heparan sulfate (HS) mimetic with low anticoagulant activity and strong potency against angiogenic growth factors and heparanase [1]. PG545 prevents neovascularization by inhibiting growth-factor HS binding and metastasis through the inhibition of HS cleavage by heparanase. This dual approach that targets both angiogenesis and heparanase has shown some clinical success with PI-88, Progen's first-in-class HS mimetic, currently in Phase II trials for melanoma. Additionally, clinical proof-of-concept with PI-88 was confirmed in Phase II trials for post-resection hepatocellular liver cancer [2], suggesting the utility of HS mimetics as anticancer agents. We have previously reported that, while PG545 and PI-88 potently inhibit experimental metastases in the B16 melanoma model, only PG545 can significantly slow solid tumor progression [1]. New tumor efficacy data and preliminary pharmacokinetic data are presented. A tumor growth inhibition (TGI) greater than 50% is considered efficacious [3].

Once- and Twice-weekly Dosing in MDA-MB-231 Xenograft Model

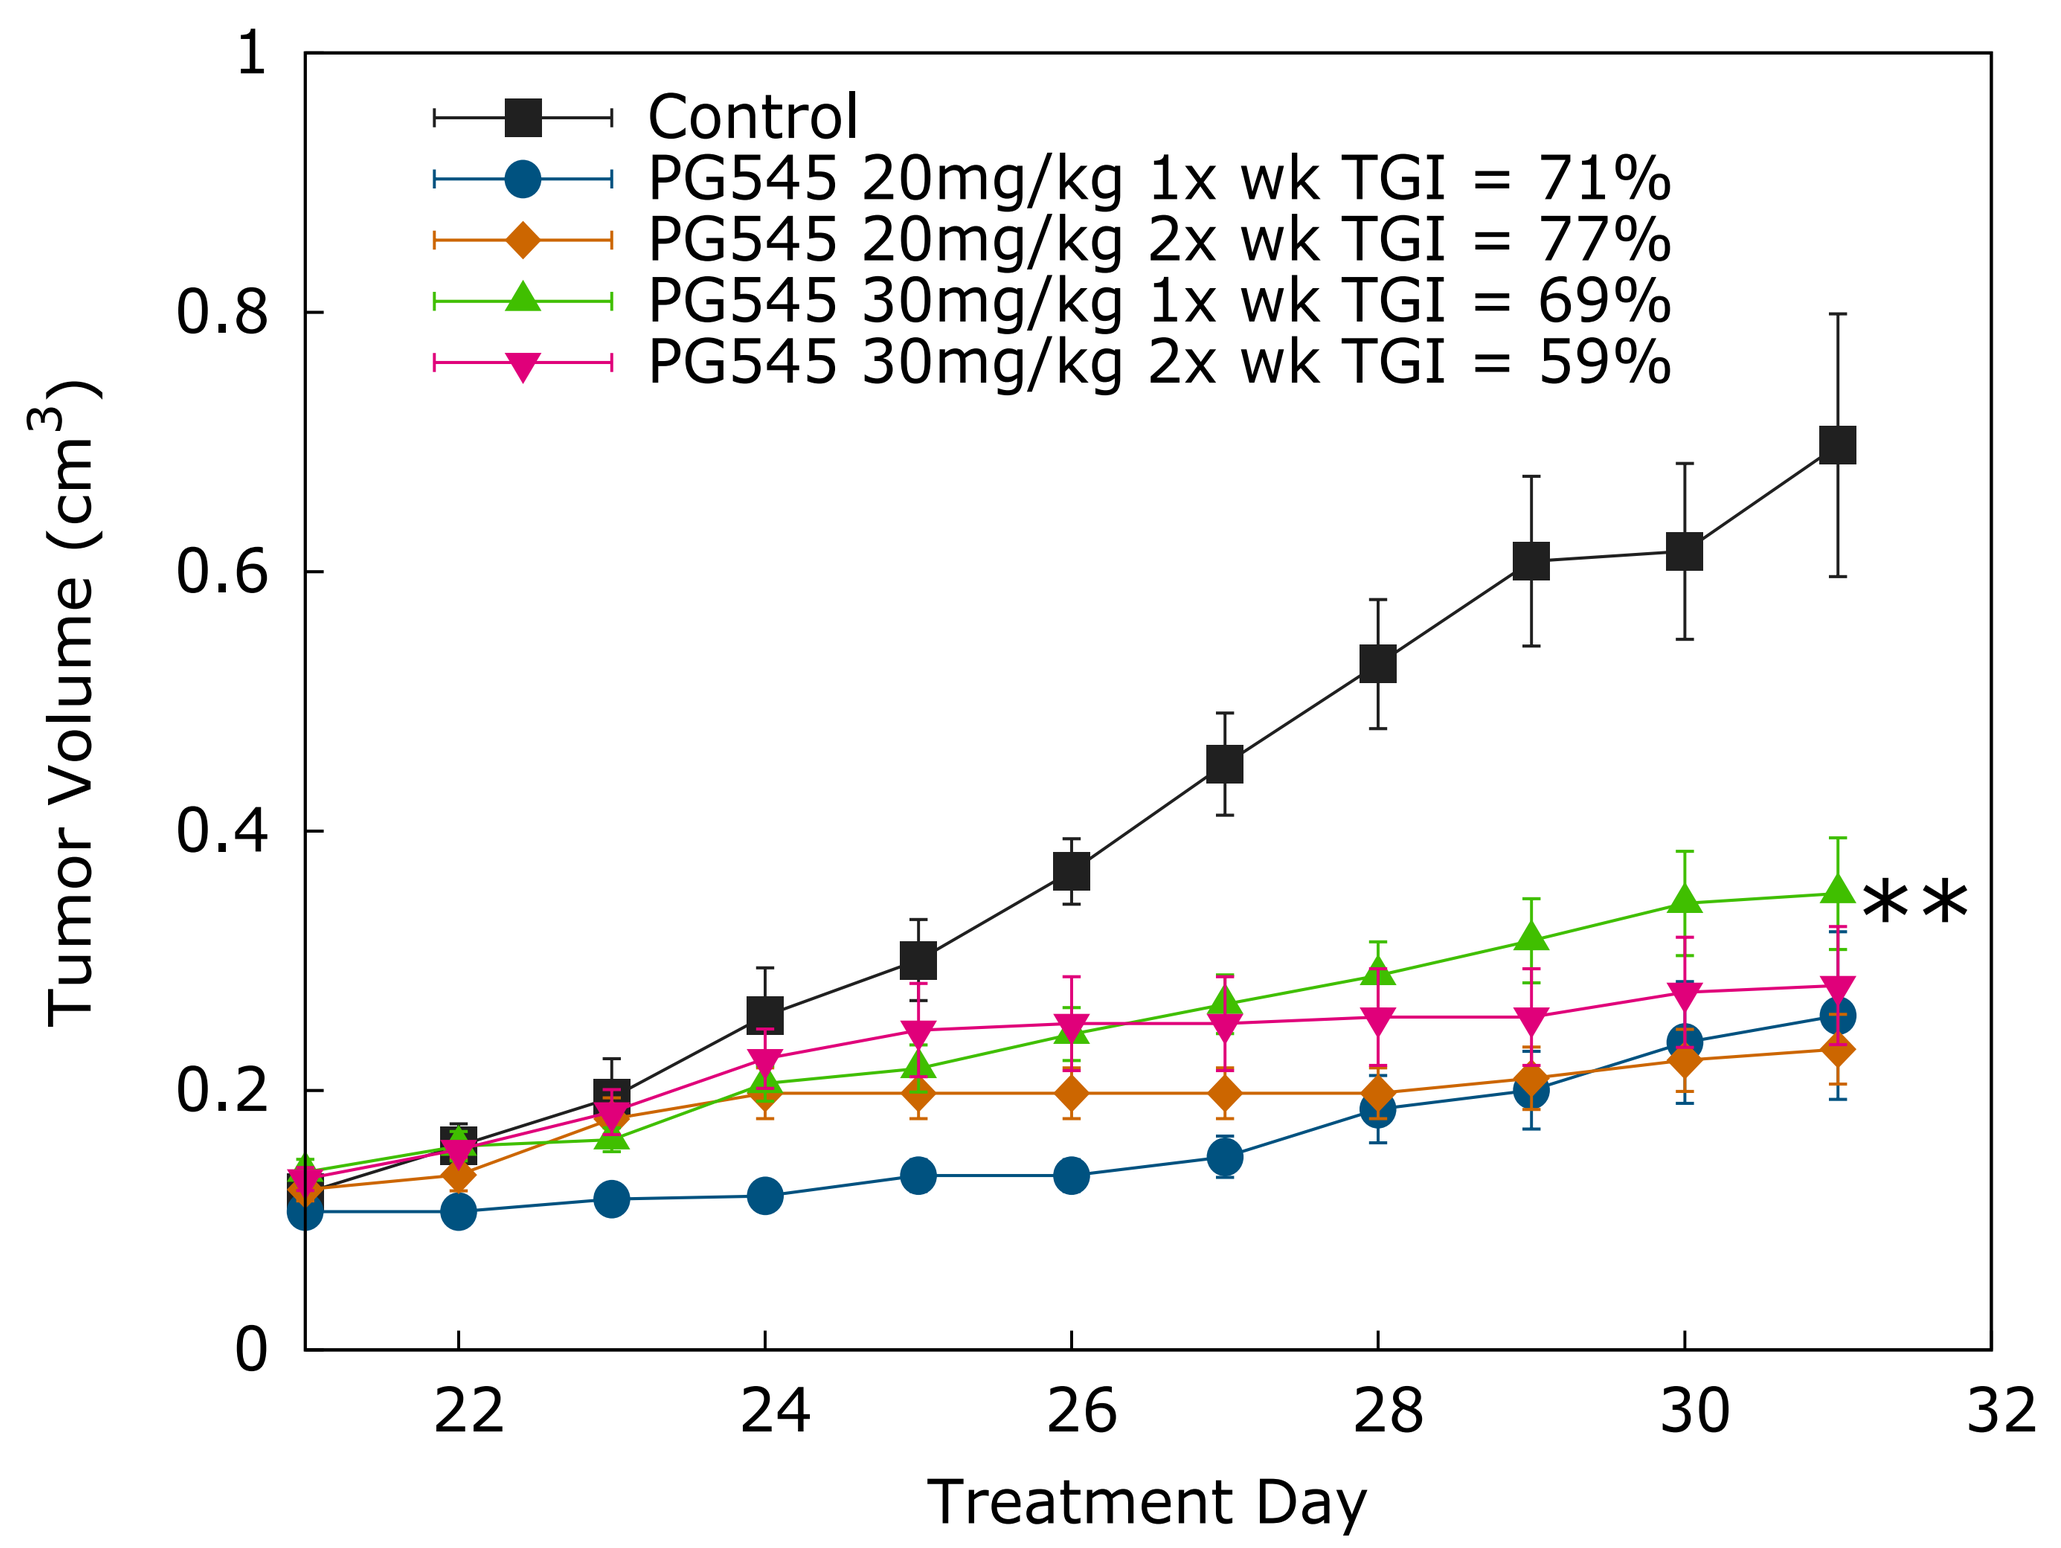
Control
PG545 20mg/kg 1x wk TGI = 71%
PG545 20mg/kg 2x wk TGI = 77%
PG545 30mg/kg 1x wk TGI = 69%
PG545 30mg/kg 2x wk TGI = 59%
Tumor Volume (cm^3)
Treatment Day
**

Figure 1
PG545 is efficacious in the MDA-MB-231 breast tumor model when dosed once- or twice-weekly. Nude mice (n=8) were injected subcutaneously with 4 x 10^6 MDA-MB-231 cells in 50 µL PBS / 50 µL Matrigel. The treatment of mice began once tumors reached a volume approximately 100mm³. Mice treated with PG545 had significantly smaller tumors versus vehicle control. **= P>0.01 versus vehicle control (repeated measures ANOVA followed by Dunnett's *post hoc* test). Bodyweight losses over the course of the study for the 20 mg/kg dose were 4% and 6% for once- and twice-weekly administration, respectively. Bodyweight losses over the course of the study for the 30 mg/kg dose were 10% and 11% for once- and twice-weekly administration, respectively.

Once- and Twice-weekly Dosing in PC3 Prostate Xenograft Model

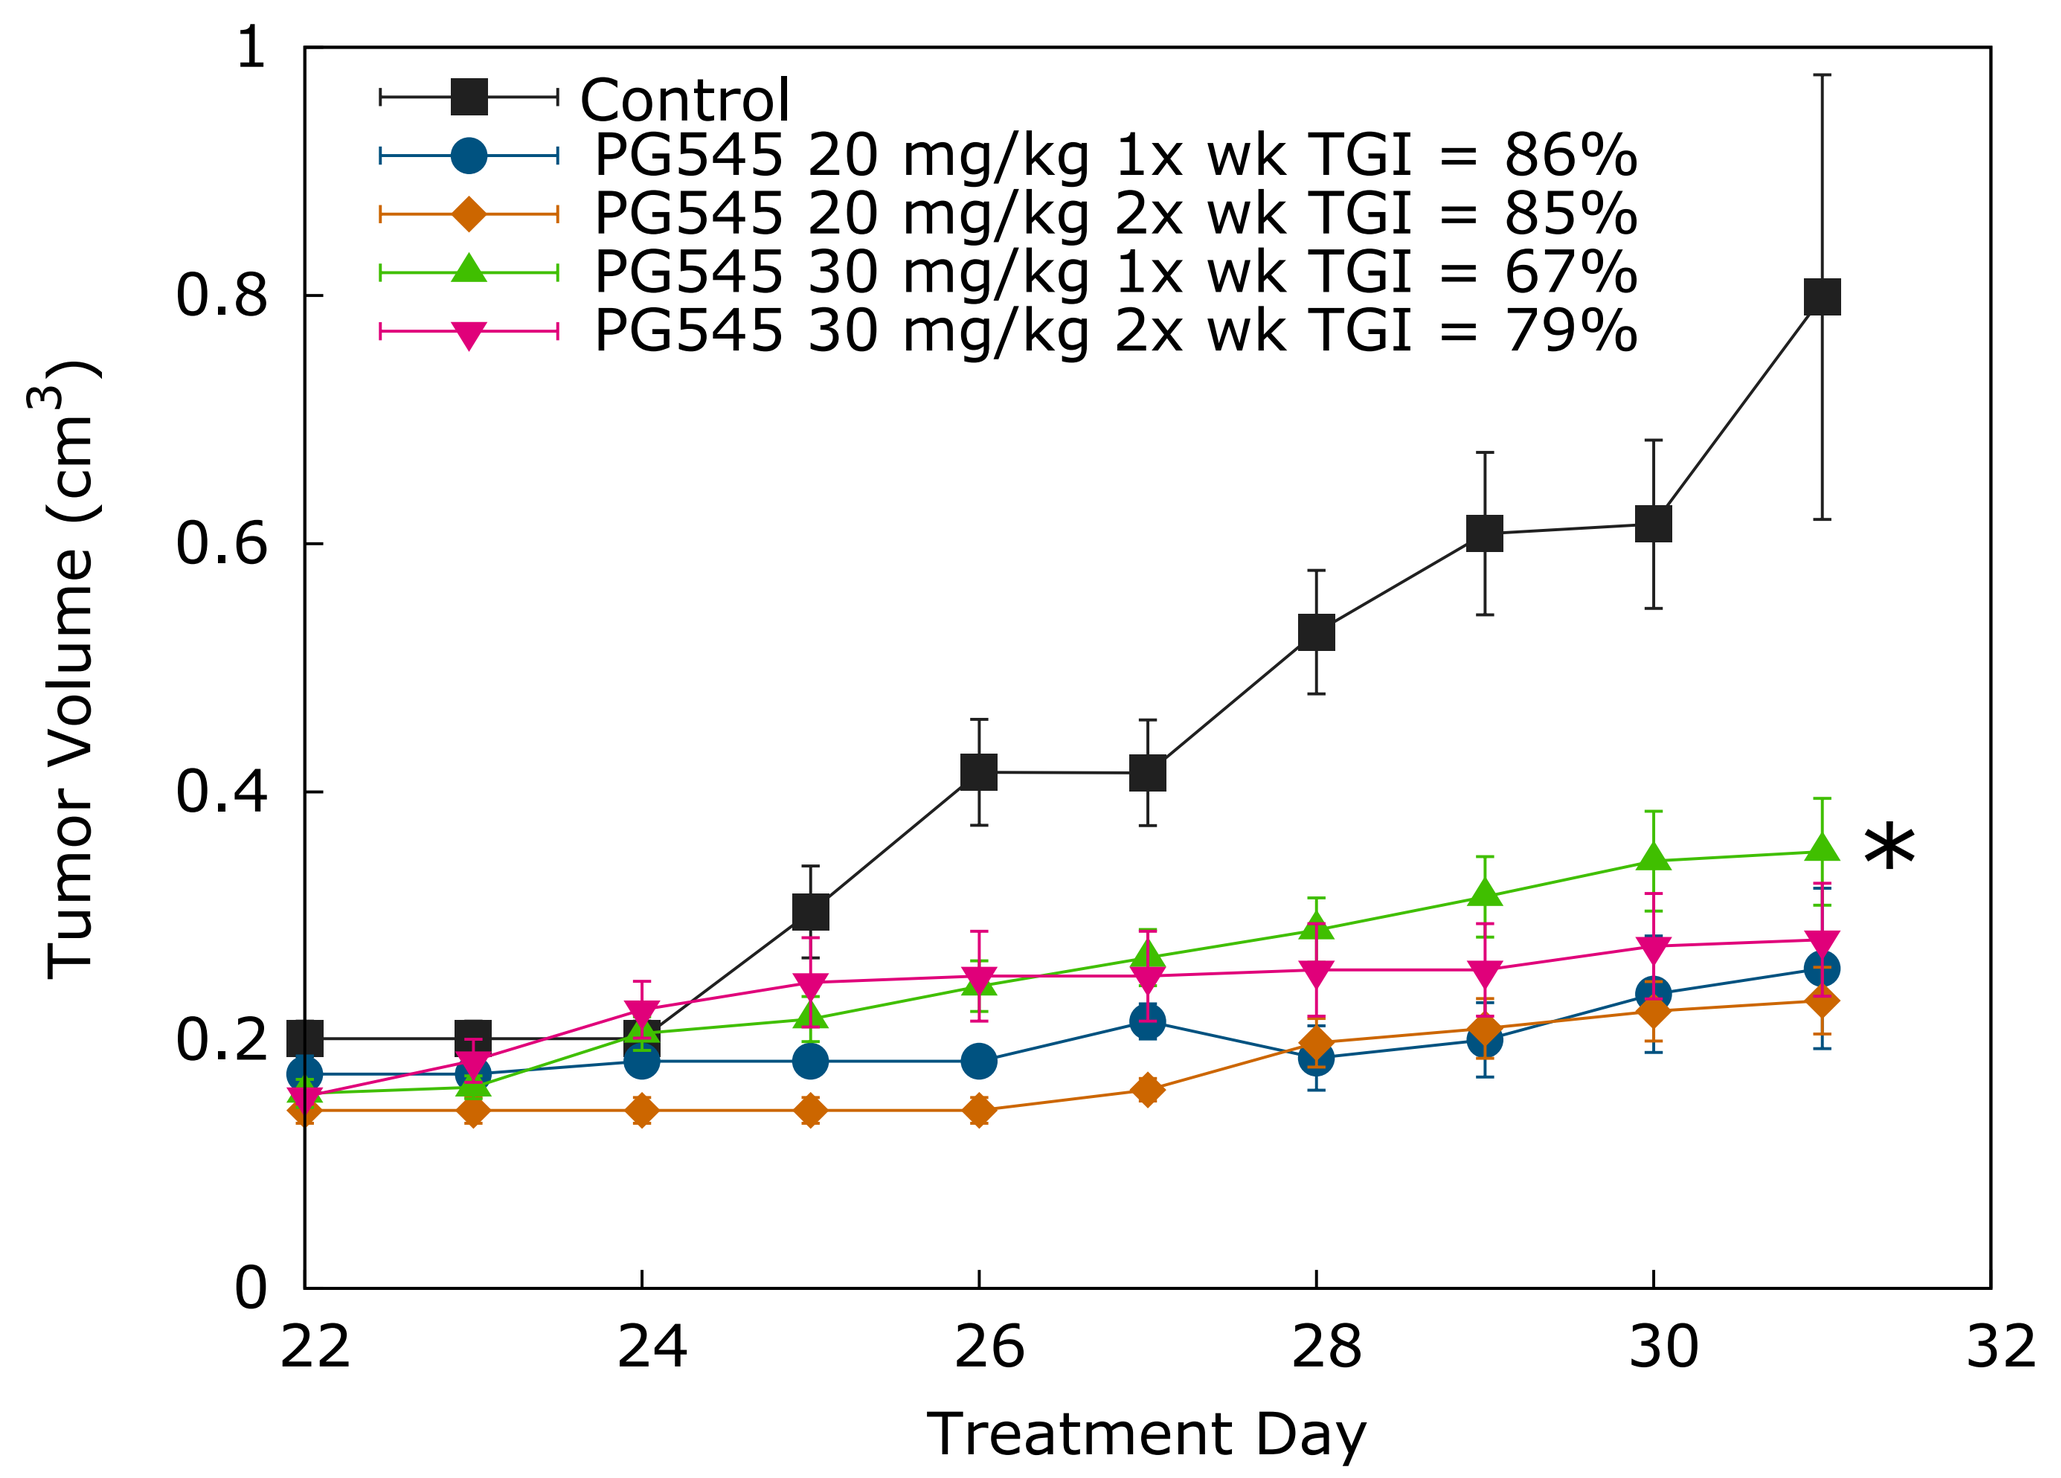
Control
PG545 20 mg/kg 1x wk TGI = 86%
PG545 20 mg/kg 2x wk TGI = 85%
PG545 30 mg/kg 1x wk TGI = 67%
PG545 30 mg/kg 2x wk TGI = 79%
Tumor Volume (cm^3)
Treatment Day
*

Figure 2
PG545 is efficacious in the PC3 prostate cancer model when dosed once- or twice-weekly. Nude mice (n=8) were injected subcutaneously with 2 x 10^6 PC3 cells in 50 µL PBS / 50 µL Matrigel. The treatment of mice began once tumors reached a volume >100mm³. Mice treated with PG545 had significantly smaller tumors versus vehicle control. **= P>0.01 versus vehicle control (repeated measures ANOVA followed by Dunnett's *post hoc* test). Bodyweight losses ranged from 6% for the once-weekly 20 mg/kg dose, to greater than 10% for the other doses.

Once- and Twice-weekly Dosing in HepG2 Liver Xenograft Model

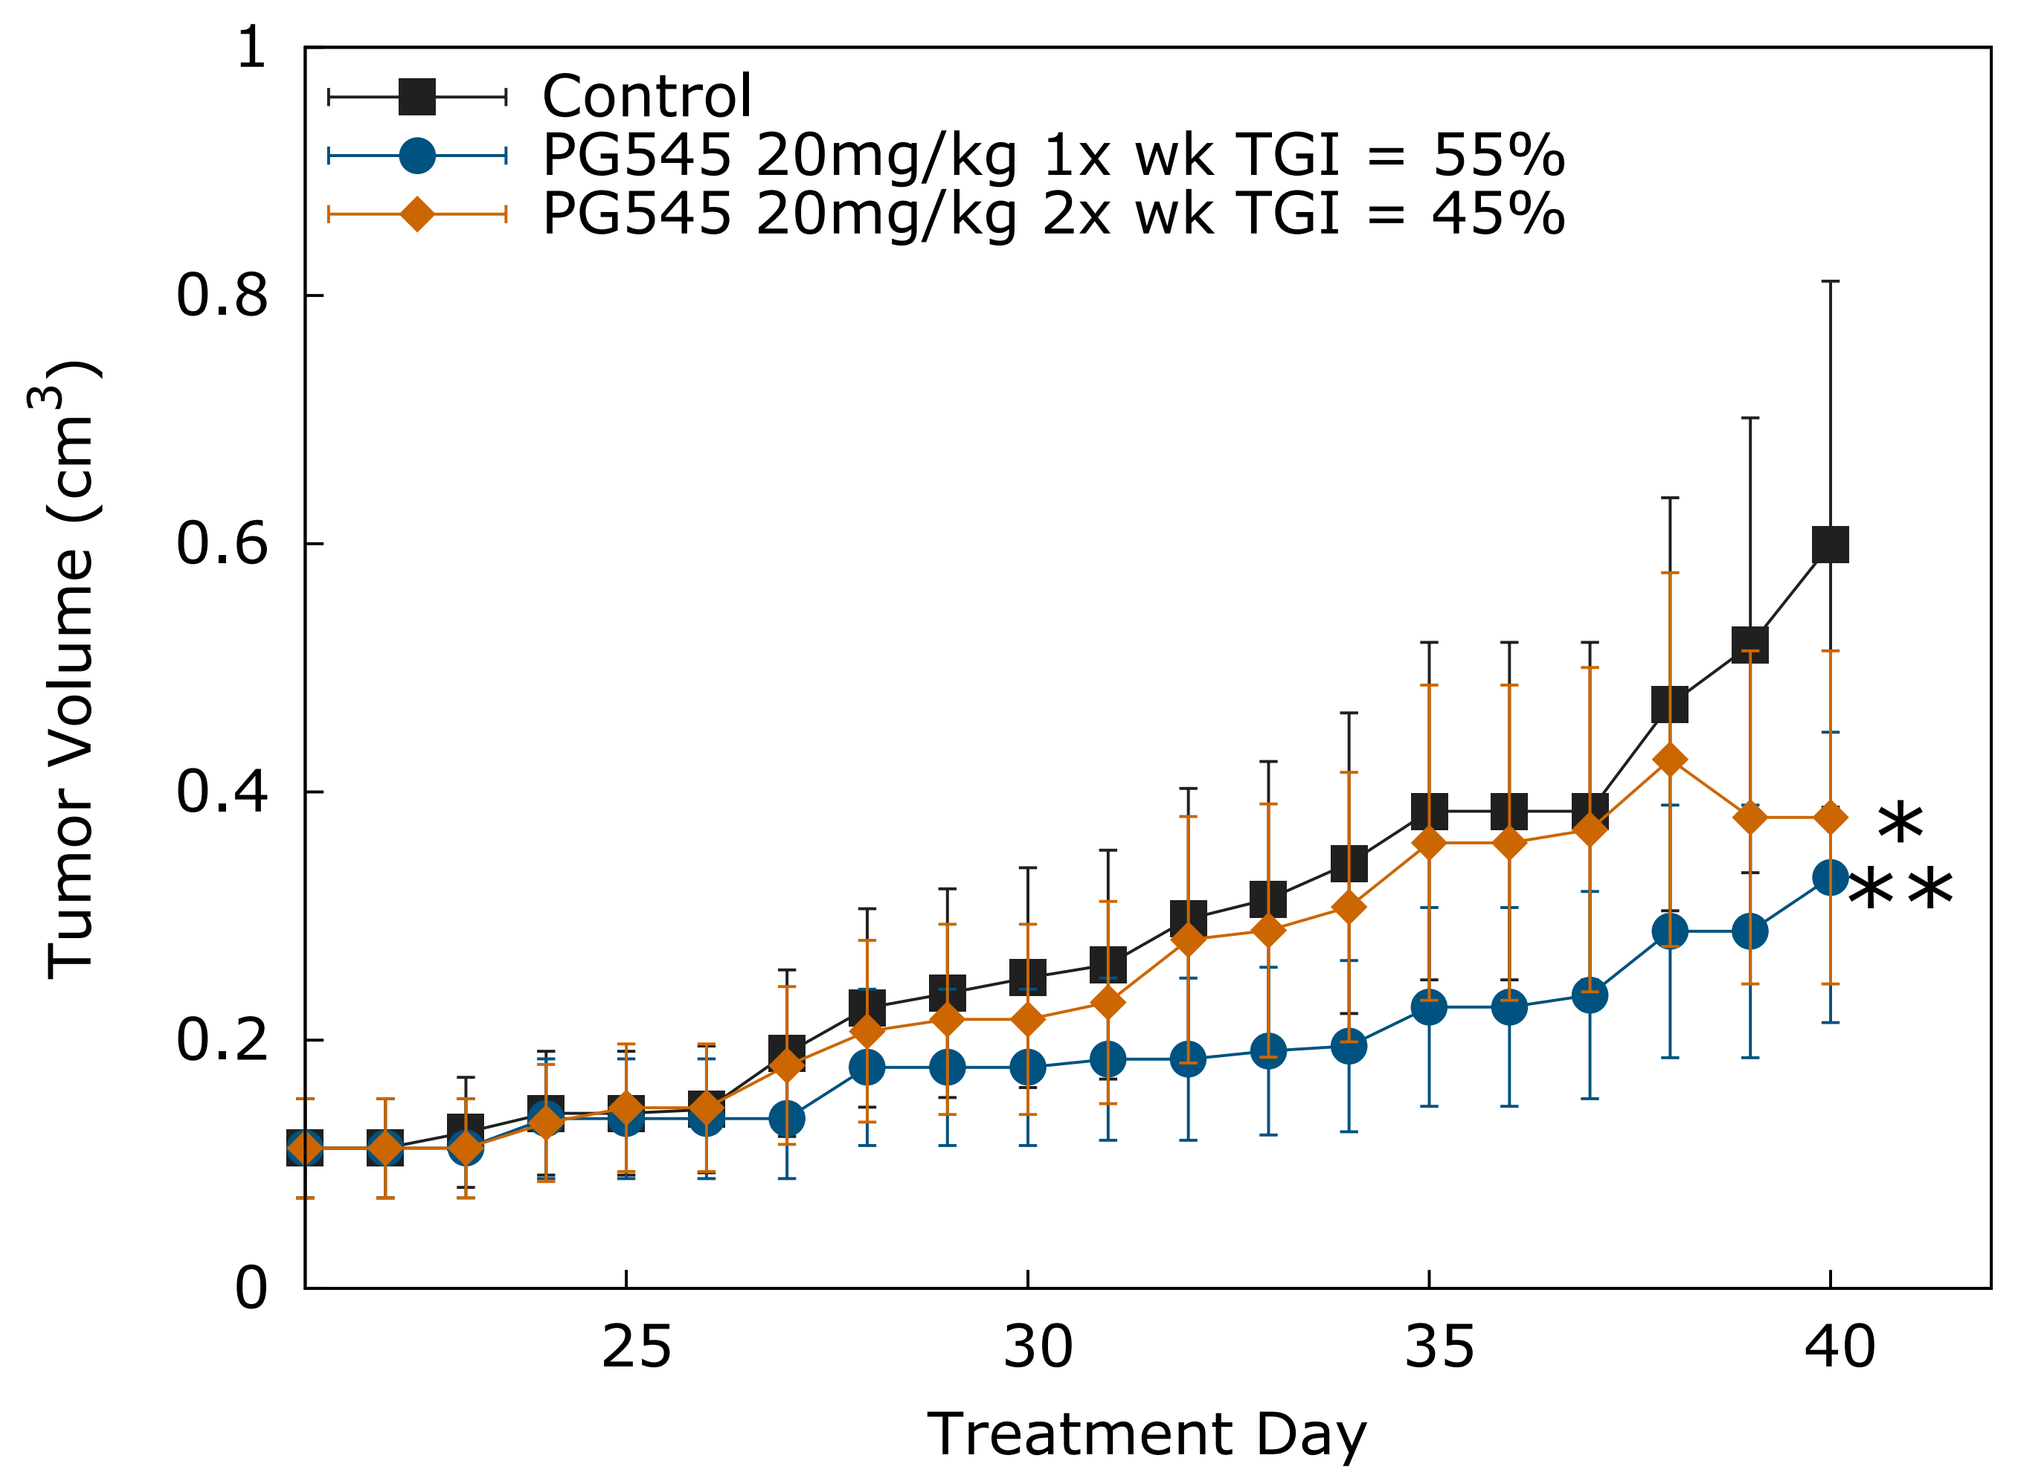
Control
PG545 20mg/kg 1x wk TGI = 55%
PG545 20mg/kg 2x wk TGI = 45%
Tumor Volume (cm^3)
Treatment Day
*
**

Figure 3
PG545 is efficacious in the HepG2 liver tumor model when dosed once- or twice-weekly. Nude mice (n=8) were injected subcutaneously with 5 x 10^5 HepG2 cells in 50 µL PBS / 50µL Matrigel. The treatment of mice began once tumors reached a volume of approximately 100mm³. At the end of treatment, mice treated with PG545 (20 mg/kg, 1x or 2x wk) had significantly smaller tumors versus vehicle control. *= P>0.05 and **=P<0.01 versus vehicle control (repeated measures ANOVA followed by Dunnett's *post hoc* test). Bodyweight losses over the course of the study were 2% and 3% for once- and twice-weekly administration, respectively.

Summary of Results

Once-weekly dosing induces significant efficacy in tumor xenograft models
- At 20 mg/kg and 30 mg/kg in the MDA-MB-231 breast cancer model model *(Figure 1)*.
- At 20 mg/kg and 30 mg/kg in the PC3 prostate cancer model *(Figure 2)*.
- At 20 mg/kg in the HepG2 liver cancer model *(Figure 3)*.
- On the basis of efficacy and limited bodyweight loss, ranging from 4% to 6%, a once-weekly dose of 20 mg/kg is considered optimal.

Hep3B orthotopic liver cancer model
- Efficacious at 20 mg/kg dosed twice-weekly with acceptable body weight loss of 9% *(Figure 4a)*.
- A further study using a loading (20 mg/kg) followed by a weekly maintenance dose (10 mg/kg) schedule also showed similar efficacy to the first model, with limited bodyweight loss ranging from 3% to 4% *(Figure 4b)*.

Pharmacokinetic properties
- Long half-life observed in blood of mice *(Figure 5a)*.
- Presence of drug in tumor support a once-weekly dosing schedule *(Figure 5b)*.

Conclusion

PG545 is efficacious against prostate, breast and liver tumors. Once-weekly dosing appears well tolerated with no significant loss of antitumor activity compared to more frequent dosing. Pharmacokinetic data in blood and tumor samples may support a once weekly dosing regimen. Together these efficacy and pharmacokinetic data support the ongoing preclinical development of PG545 as a once-weekly, subcutaneously administered injection for the treatment of cancer.

References

1. Dredge *et al. Invest New Drugs* (2009) In Press.
2. Liu *et al. Journal of Hepatology* (2009) 50 (5): 850-853.
3. Chang *et al. Cancer Chemother Pharmacol* (2007) 59: 561-574

Acknowledgements

Many thanks to Ralf Brandt and the team at vivoPharm who conducted the Hep3b2.1-7 experiment and Russell Addison at Tetra-Q for facilitation and discussion relating to the pharmacokinetic analysis.

Twice-weekly Dosing in an Orthotopic Liver Cancer Model

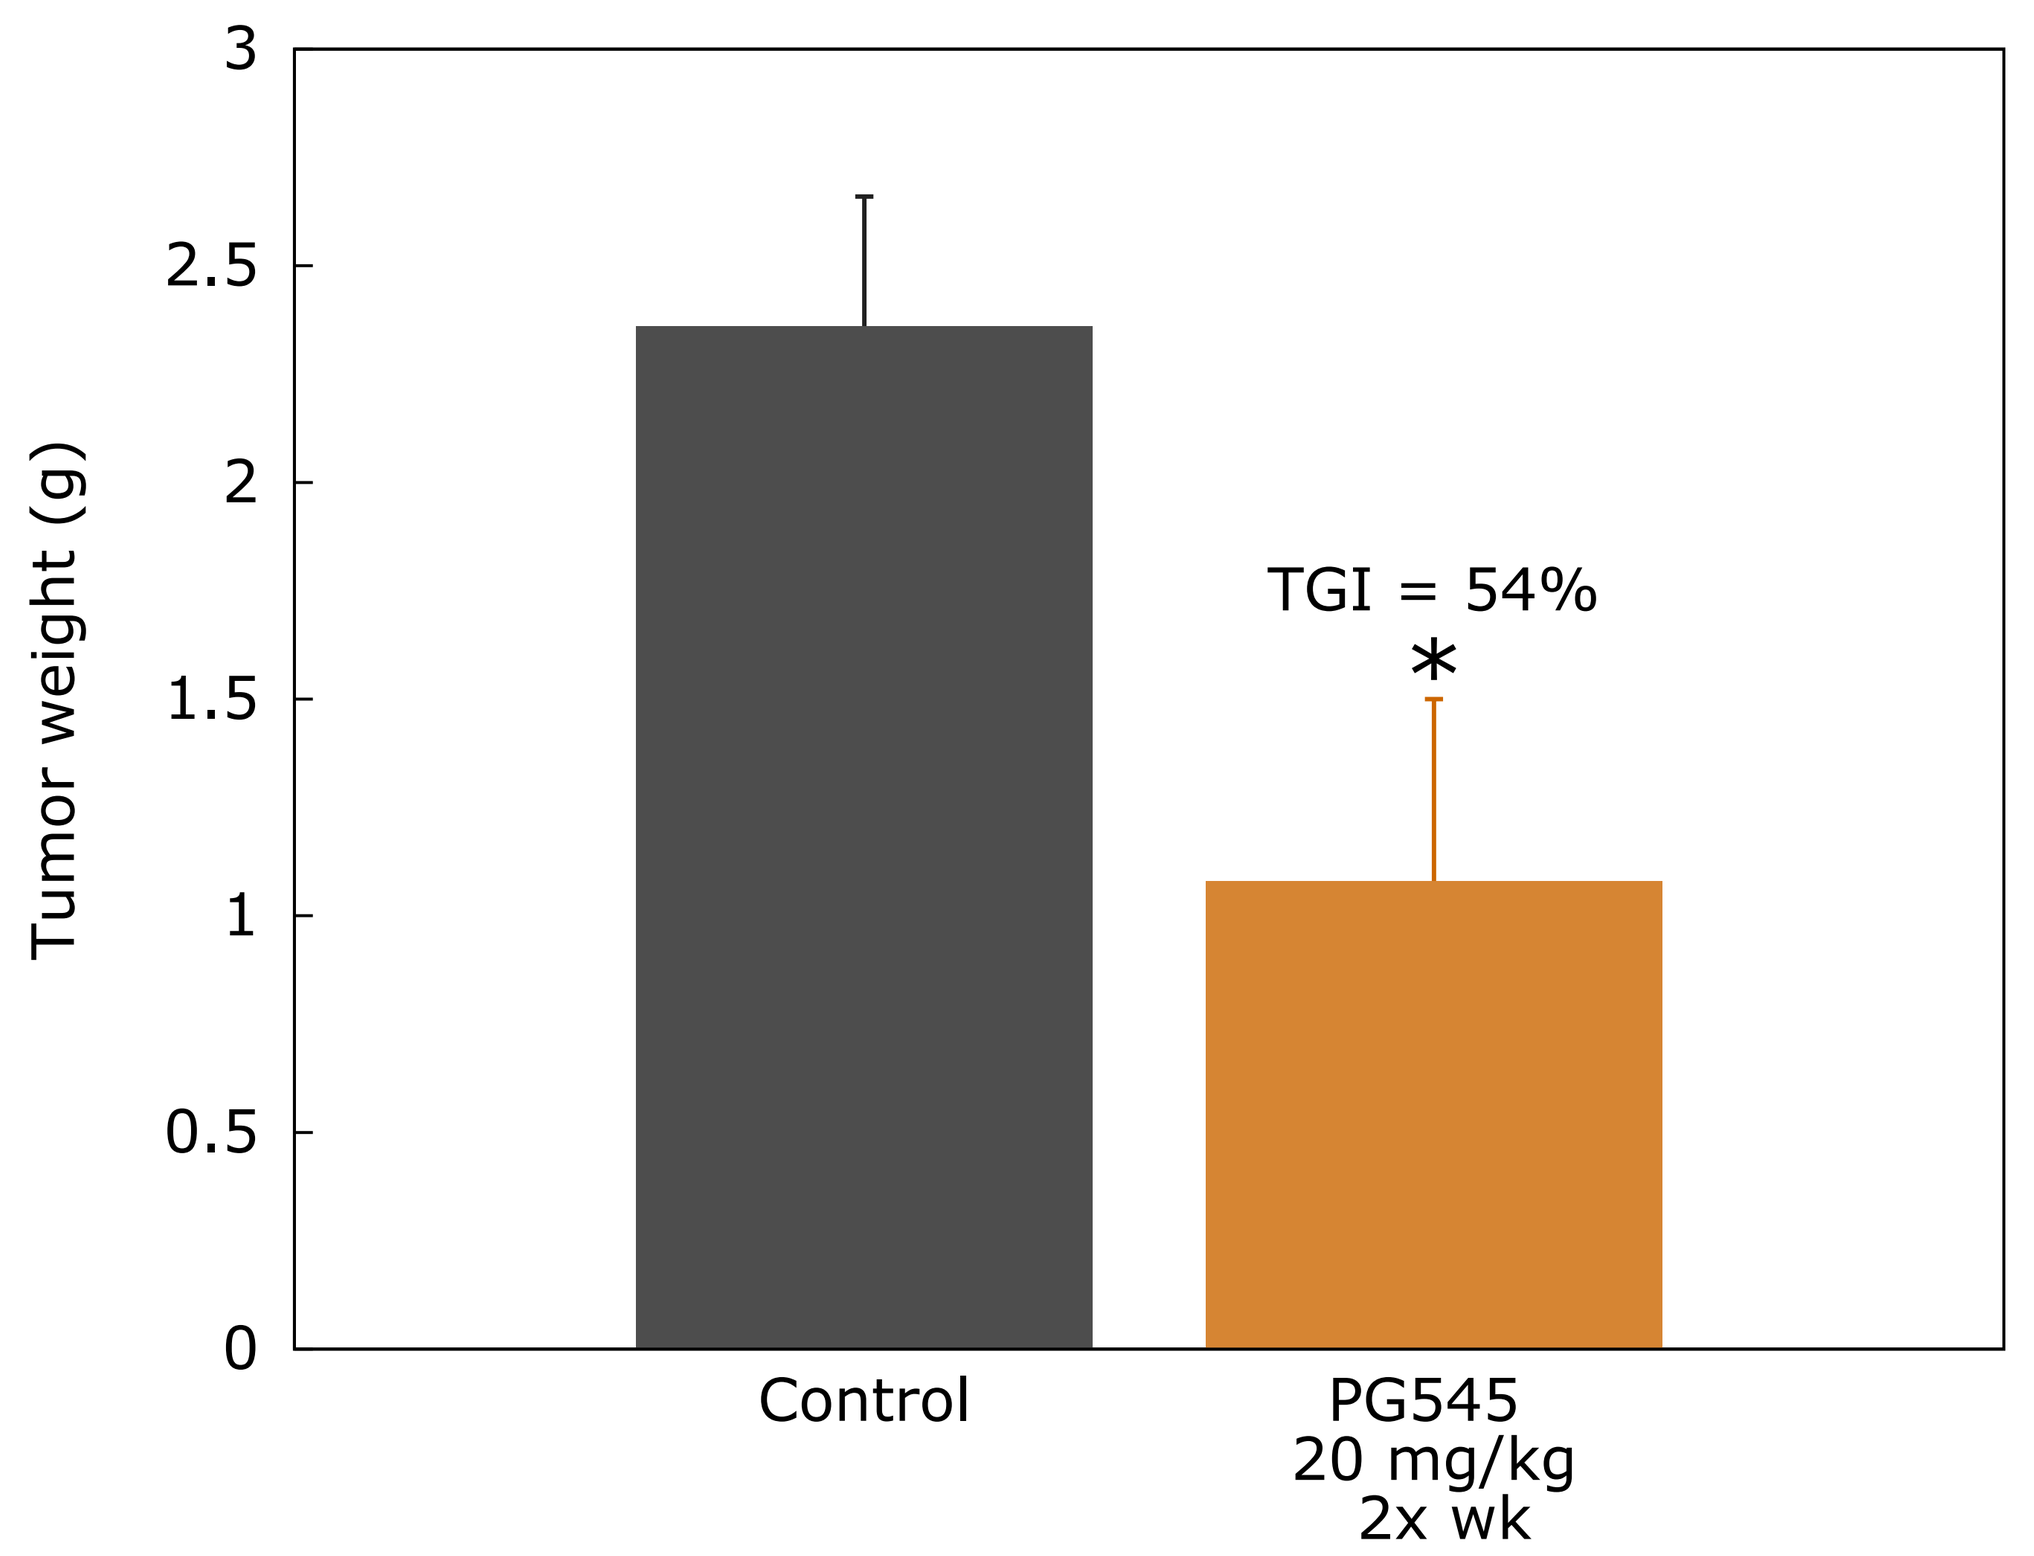
Tumor weight (g)
TGI = 54%
*
Control
PG545
20 mg/kg
2x wk

Figure 4a
PG545 is efficacious in the Hep3B2 hepatocellular carcinoma model when dosed twice-weekly at 20 mg/kg. SCID mice (n=6-7) were injected orthotopically with 2 x 10^6 Hep3b2.1-7 cells in 10 µL with 50% Matrigel. The treatment of mice began 21 days after inoculation of the Hep3B2.1-7 cells. Mice treated with 20 mg/kg PG545 had smaller tumors upon study termination compared to those treated with the Vehicle Control. Historical data from the same laboratory found a similar TGI using Sorafenib at 30 mg/kg. *=P>0.05 versus vehicle control (Unpaired t-test). Bodyweight loss over the course of treatment was 9%.

[^{3}H]-PG545: PK Profile in Blood

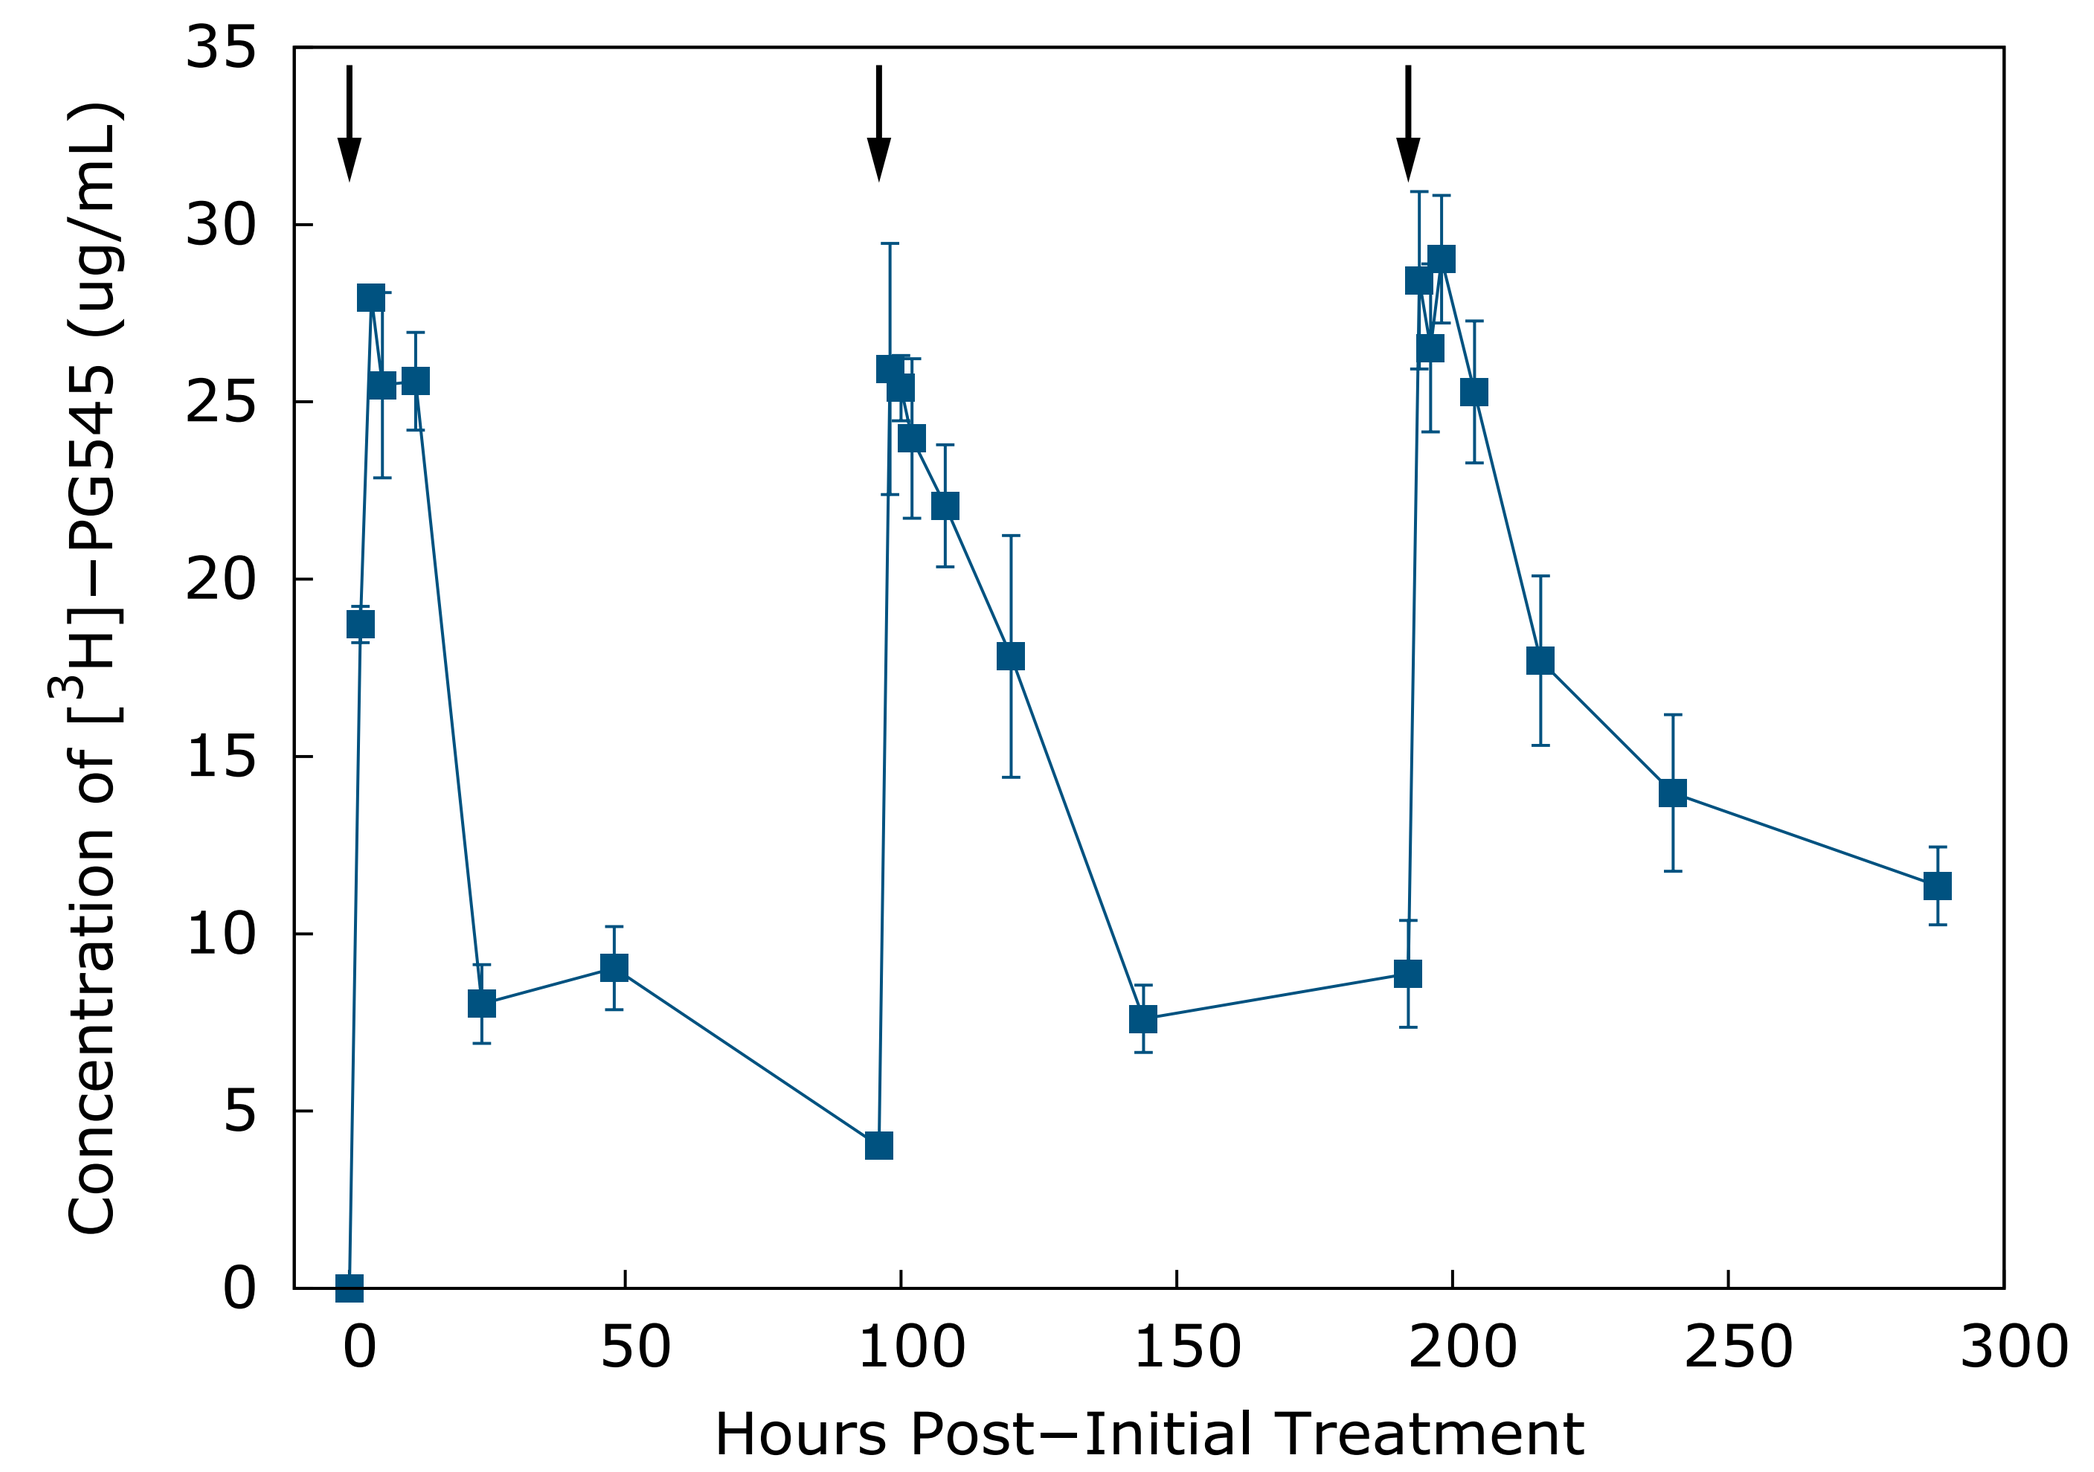
Concentration of [^{3}H]−PG545 (ug/mL)
Hours Post−Initial Treatment

Figure 5a
Concentration of [^{3}H]-PG545 in plasma. The observed plasma concentration of [^{3}H]-PG545 over time following administration of three 20 mg/kg (SC) bolus doses of [^{3}H]-PG545 to groups of tumor-bearing mice is shown. Each dose, denoted by the arrow, was separated by 96 h.

Once-weekly Loading and Maintenance Dose Schedule is Efficacious in an Orthotopic Liver Cancer Model

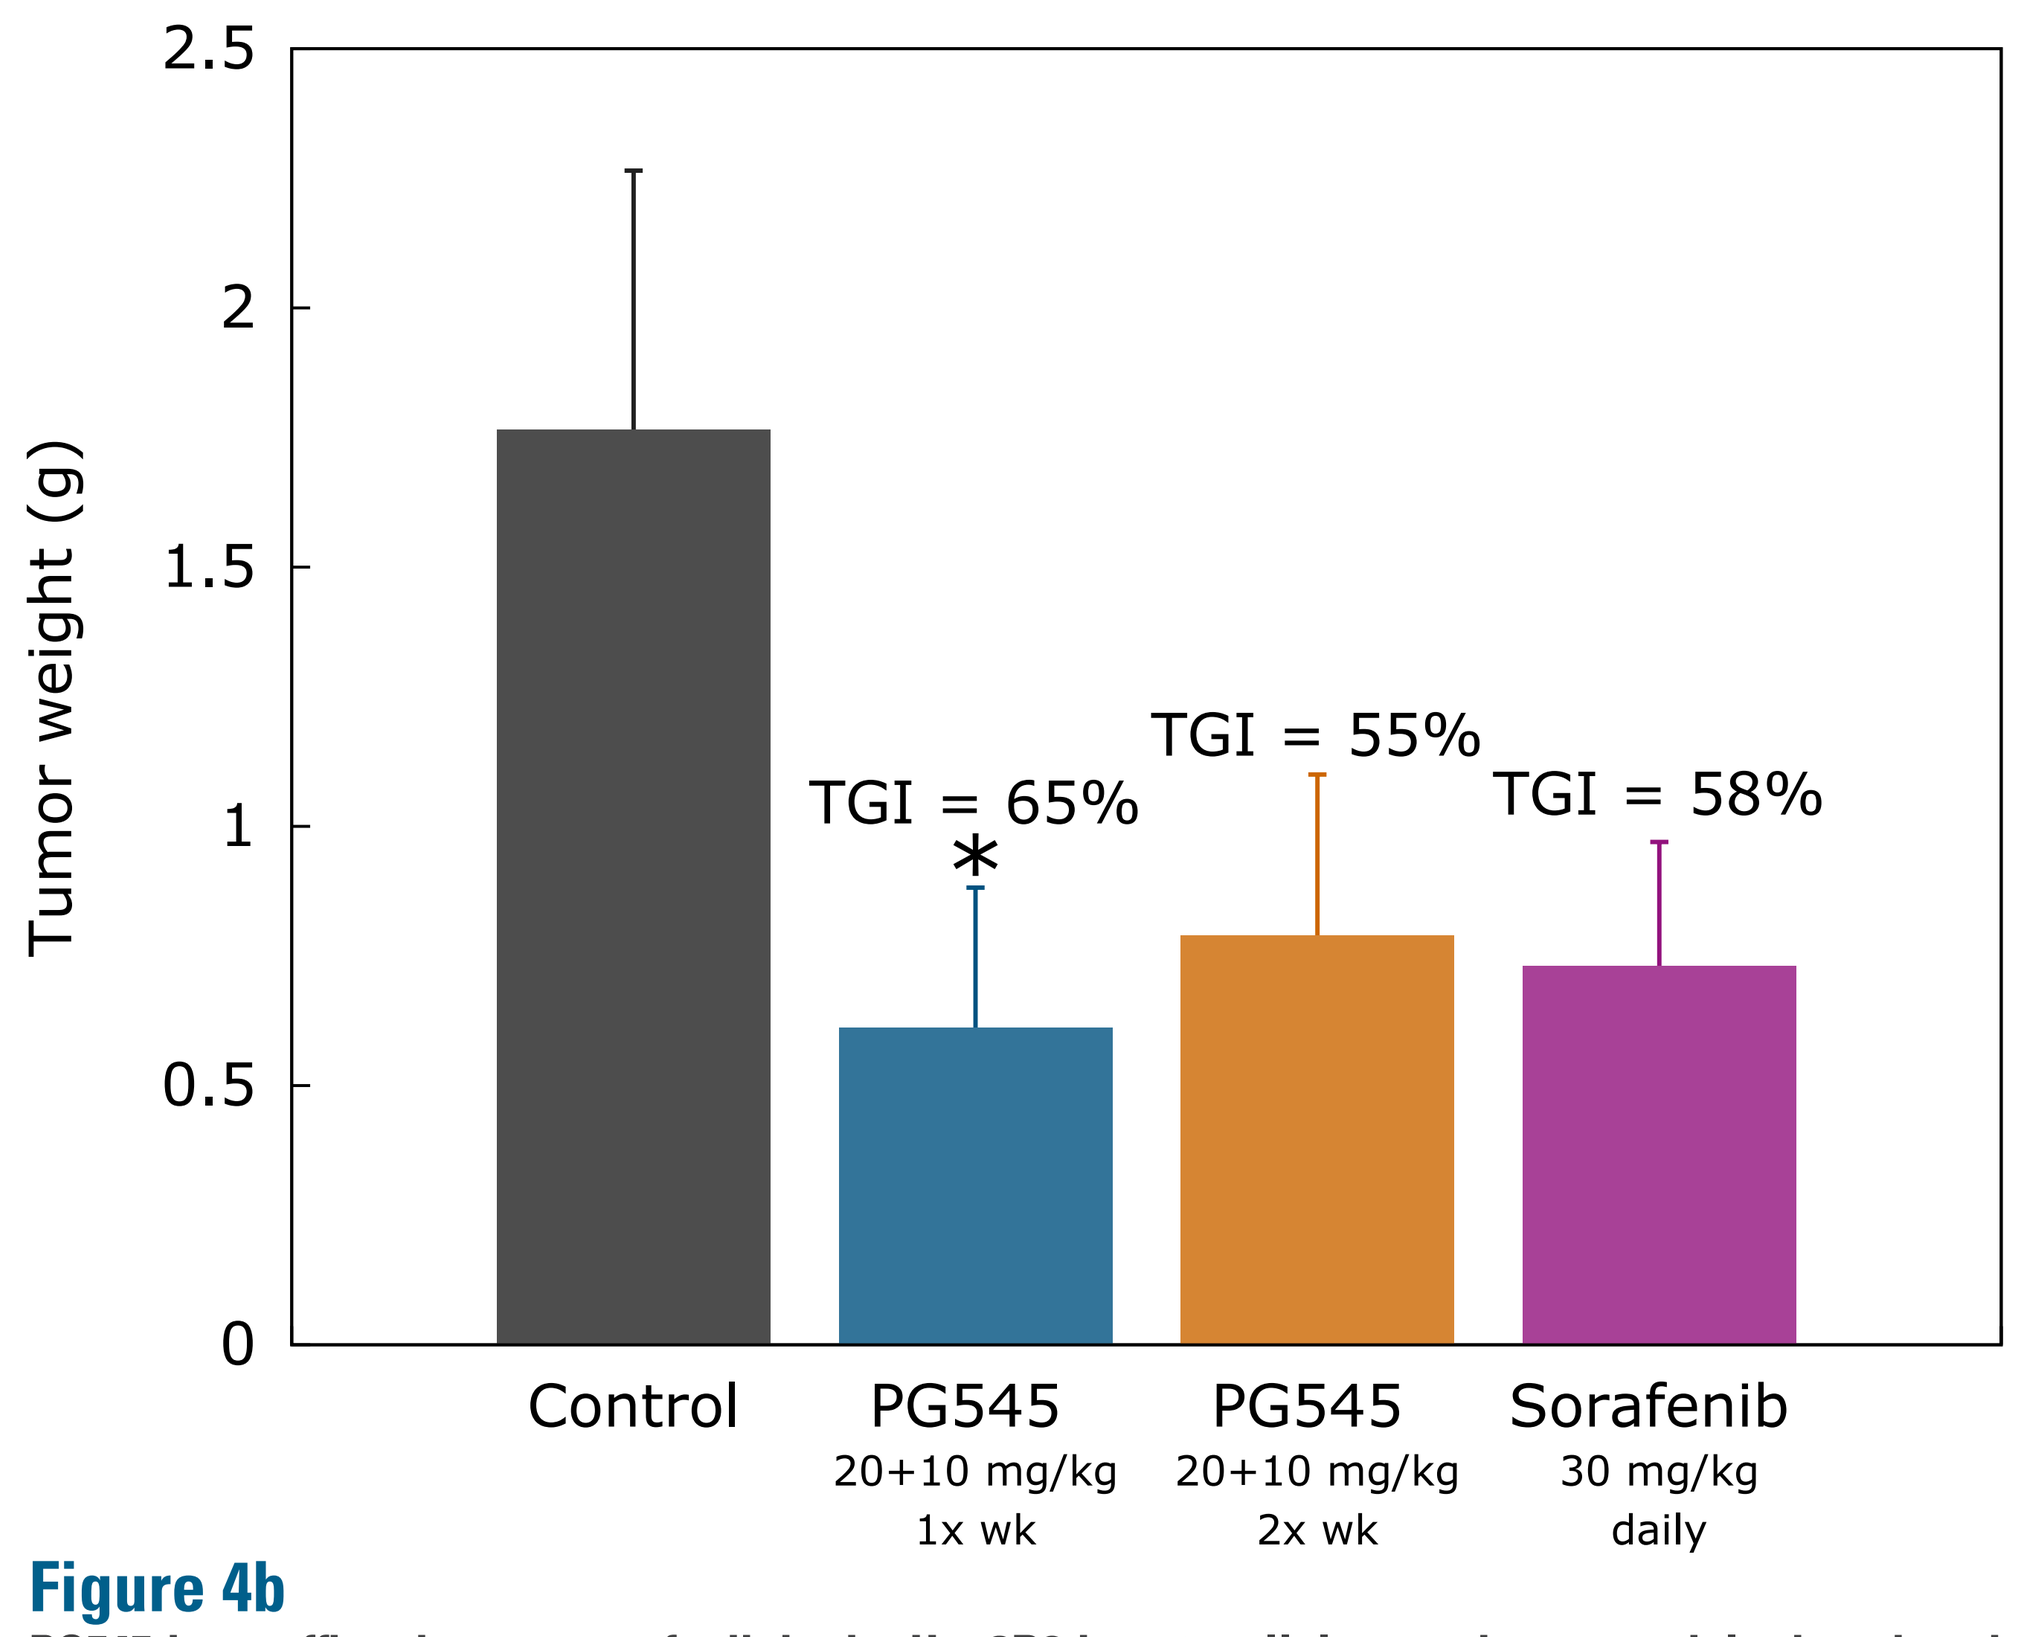
Tumor weight (g)
TGI = 65%
*
TGI = 55%
TGI = 58%
Control
PG545
20+10 mg/kg
1x wk
PG545
20+10 mg/kg
2x wk
Sorafenib
30 mg/kg
daily

Figure 4b
PG545 is as efficacious as sorafenib in the Hep3B2 hepatocellular carcinoma model when dosed once- and twice-weekly. SCID mice (n=6-7) were injected orthotopically with 2 x 10^6 Hep3b2.1-7 cells in 10 µL with 50% Matrigel. The treatment of mice began 21 days after inoculation of the Hep3B2.1-7 cells. Mice treated with a loading dose of 20 mg/kg PG545 followed by 1x or 2x weekly injections of 10 mg/kg for two weeks. Upon study termination, PG545 and sorafenib-treated mice had lower tumors compared to those treated with the Vehicle control. *=P>0.05 versus vehicle control (ANOVA followed by Dunnett's test). Bodyweight losses over the course of the study were 3% and 4% for once- and twice-weekly doses of PG545, respectively, and 2% for daily 30 mg/kg Sorafenib.

[^{3}H]-PG545: PK Profile in Tumor Tissue (Hep3B)

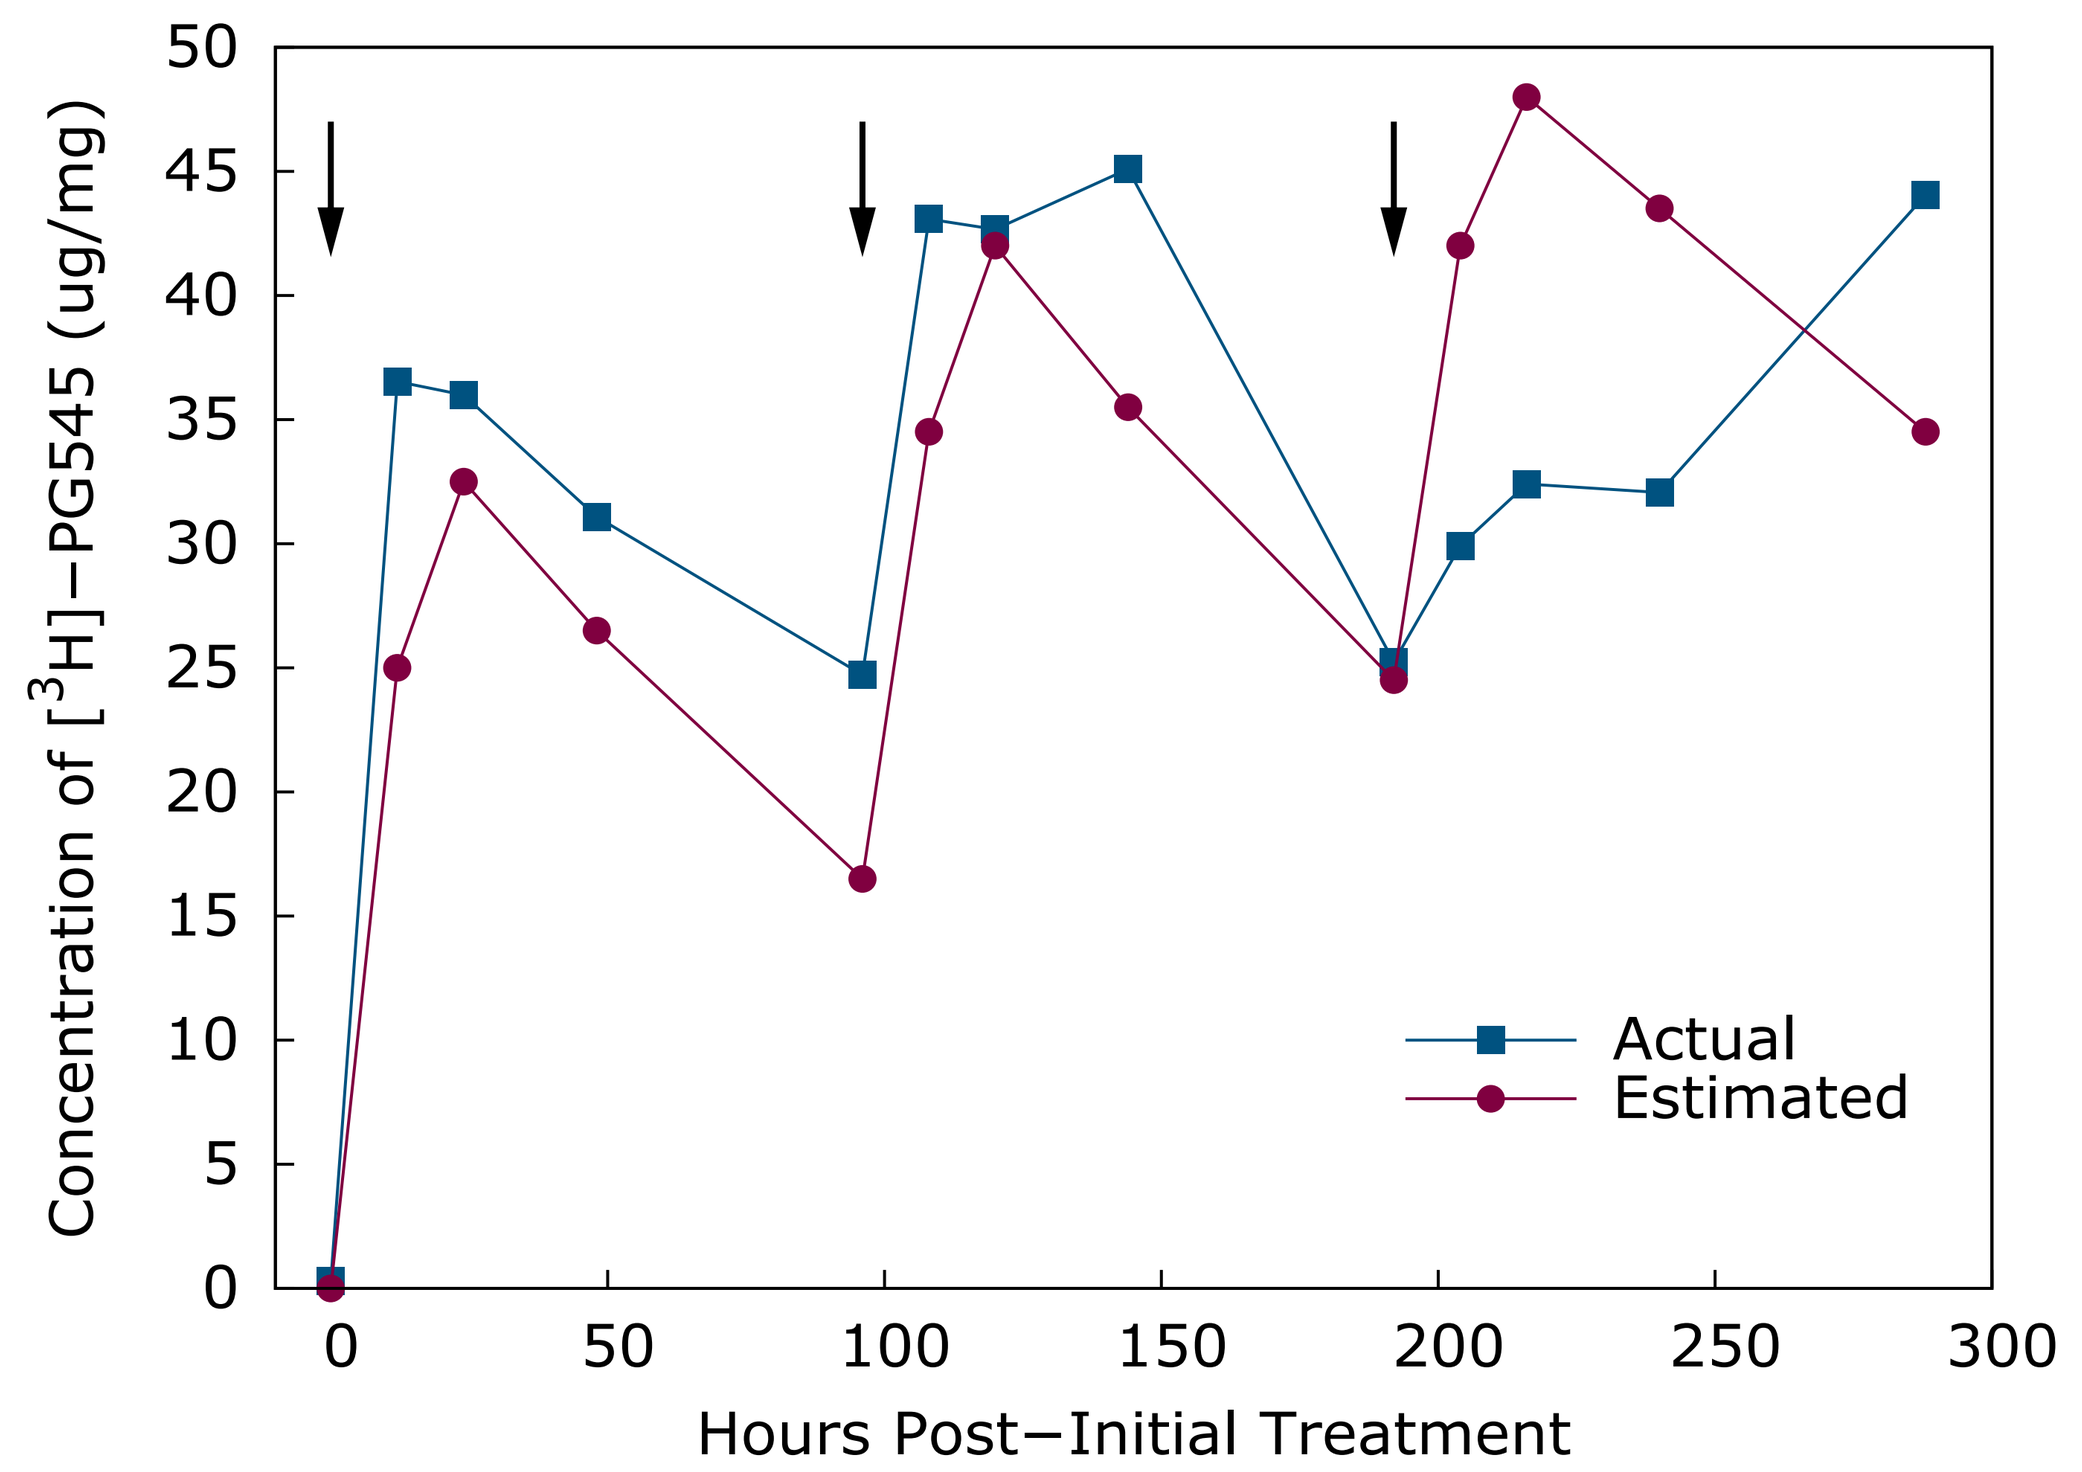
Concentration of [^{3}H]−PG545 (ug/mg)
Hours Post−Initial Treatment
Actual
Estimated

Figure 5b
Concentration of [^{3}H]-PG545 in tumor. Mean actual (blue squares) and estimated (red circles) tumor concentration versus time data for [^{3}H]-PG545 following administration of three 20 mg/kg (SC) bolus doses of [^{3}H]-PG545 to groups of tumor-bearing mice. Each dose, denoted by the arrows, was separated by 96 h. Tumor concentrations were estimated using a one compartment pharmacokinetic model such that tumor concentrations reflect perfusion of the tumor mass with blood containing [^{3}H]-PG545 and where the blood [^{3}H]-PG545 concentration was changing with time.