HEPARAN SULFATE MIMETICS – POTENT ANGIOGENESIS INHIBITORS FOR CANCER THERAPY

Dredge K, Davis K, Liu L, Johnstone K, Handley P, Wimmer N, Bytheway I, Hammond E, Li CP, Rowley J, Ferro V, Gautam A. Drug Design Group, Progen Pharmaceuticals, Brisbane, Qld, Australia.

PROGEN PHARMACEUTICALS

INTRODUCTION

- Heparan sulfate (HS) proteoglycans play critical roles in pathophysiological processes associated with tumor development such as angiogenesis and metastasis
- Several growth factors, their receptors, extracellular matrix molecules and enzymes bind to specific sites on the HS sugar chain to facilitate cellular and biochemical responses
- HS mimetics function by blocking these interactions and inhibiting processes crucial to tumor progression
- PI-88 is an example of a HS mimetic planned to undergo clinical investigation in a Phase 3 trial later this year as an adjuvant therapy for post-resection hepatocellular carcinoma. PI-88 interferes with two important processes in tumor development. PI-88 interferes with two important processes in tumor development:
 - Angiogenesis *via* inhibition of VEGF, FGF-1, FGF-2 and induction of TFPI
 - Metastasis *via* inhibition of heparanase
- Progen has developed the PG500 series program to design a HS mimetic with the following properties:
 - New chemical entity (NCE) on a single carbon backbone
 - Potent angiogenesis inhibitor
 - Potent antitumor activity
 - Low anti-coagulant activity
 - Acceptable toxicological and pharmacokinetic profile

The data presented here demonstrates the potent activity of lead PG500 series compounds in *in vitro* and *ex vivo* angiogenesis assays and *in vivo* tumor models

FGF-1, FGF-2 & VEGF-INDUCED HUVEC PROLIFERATION

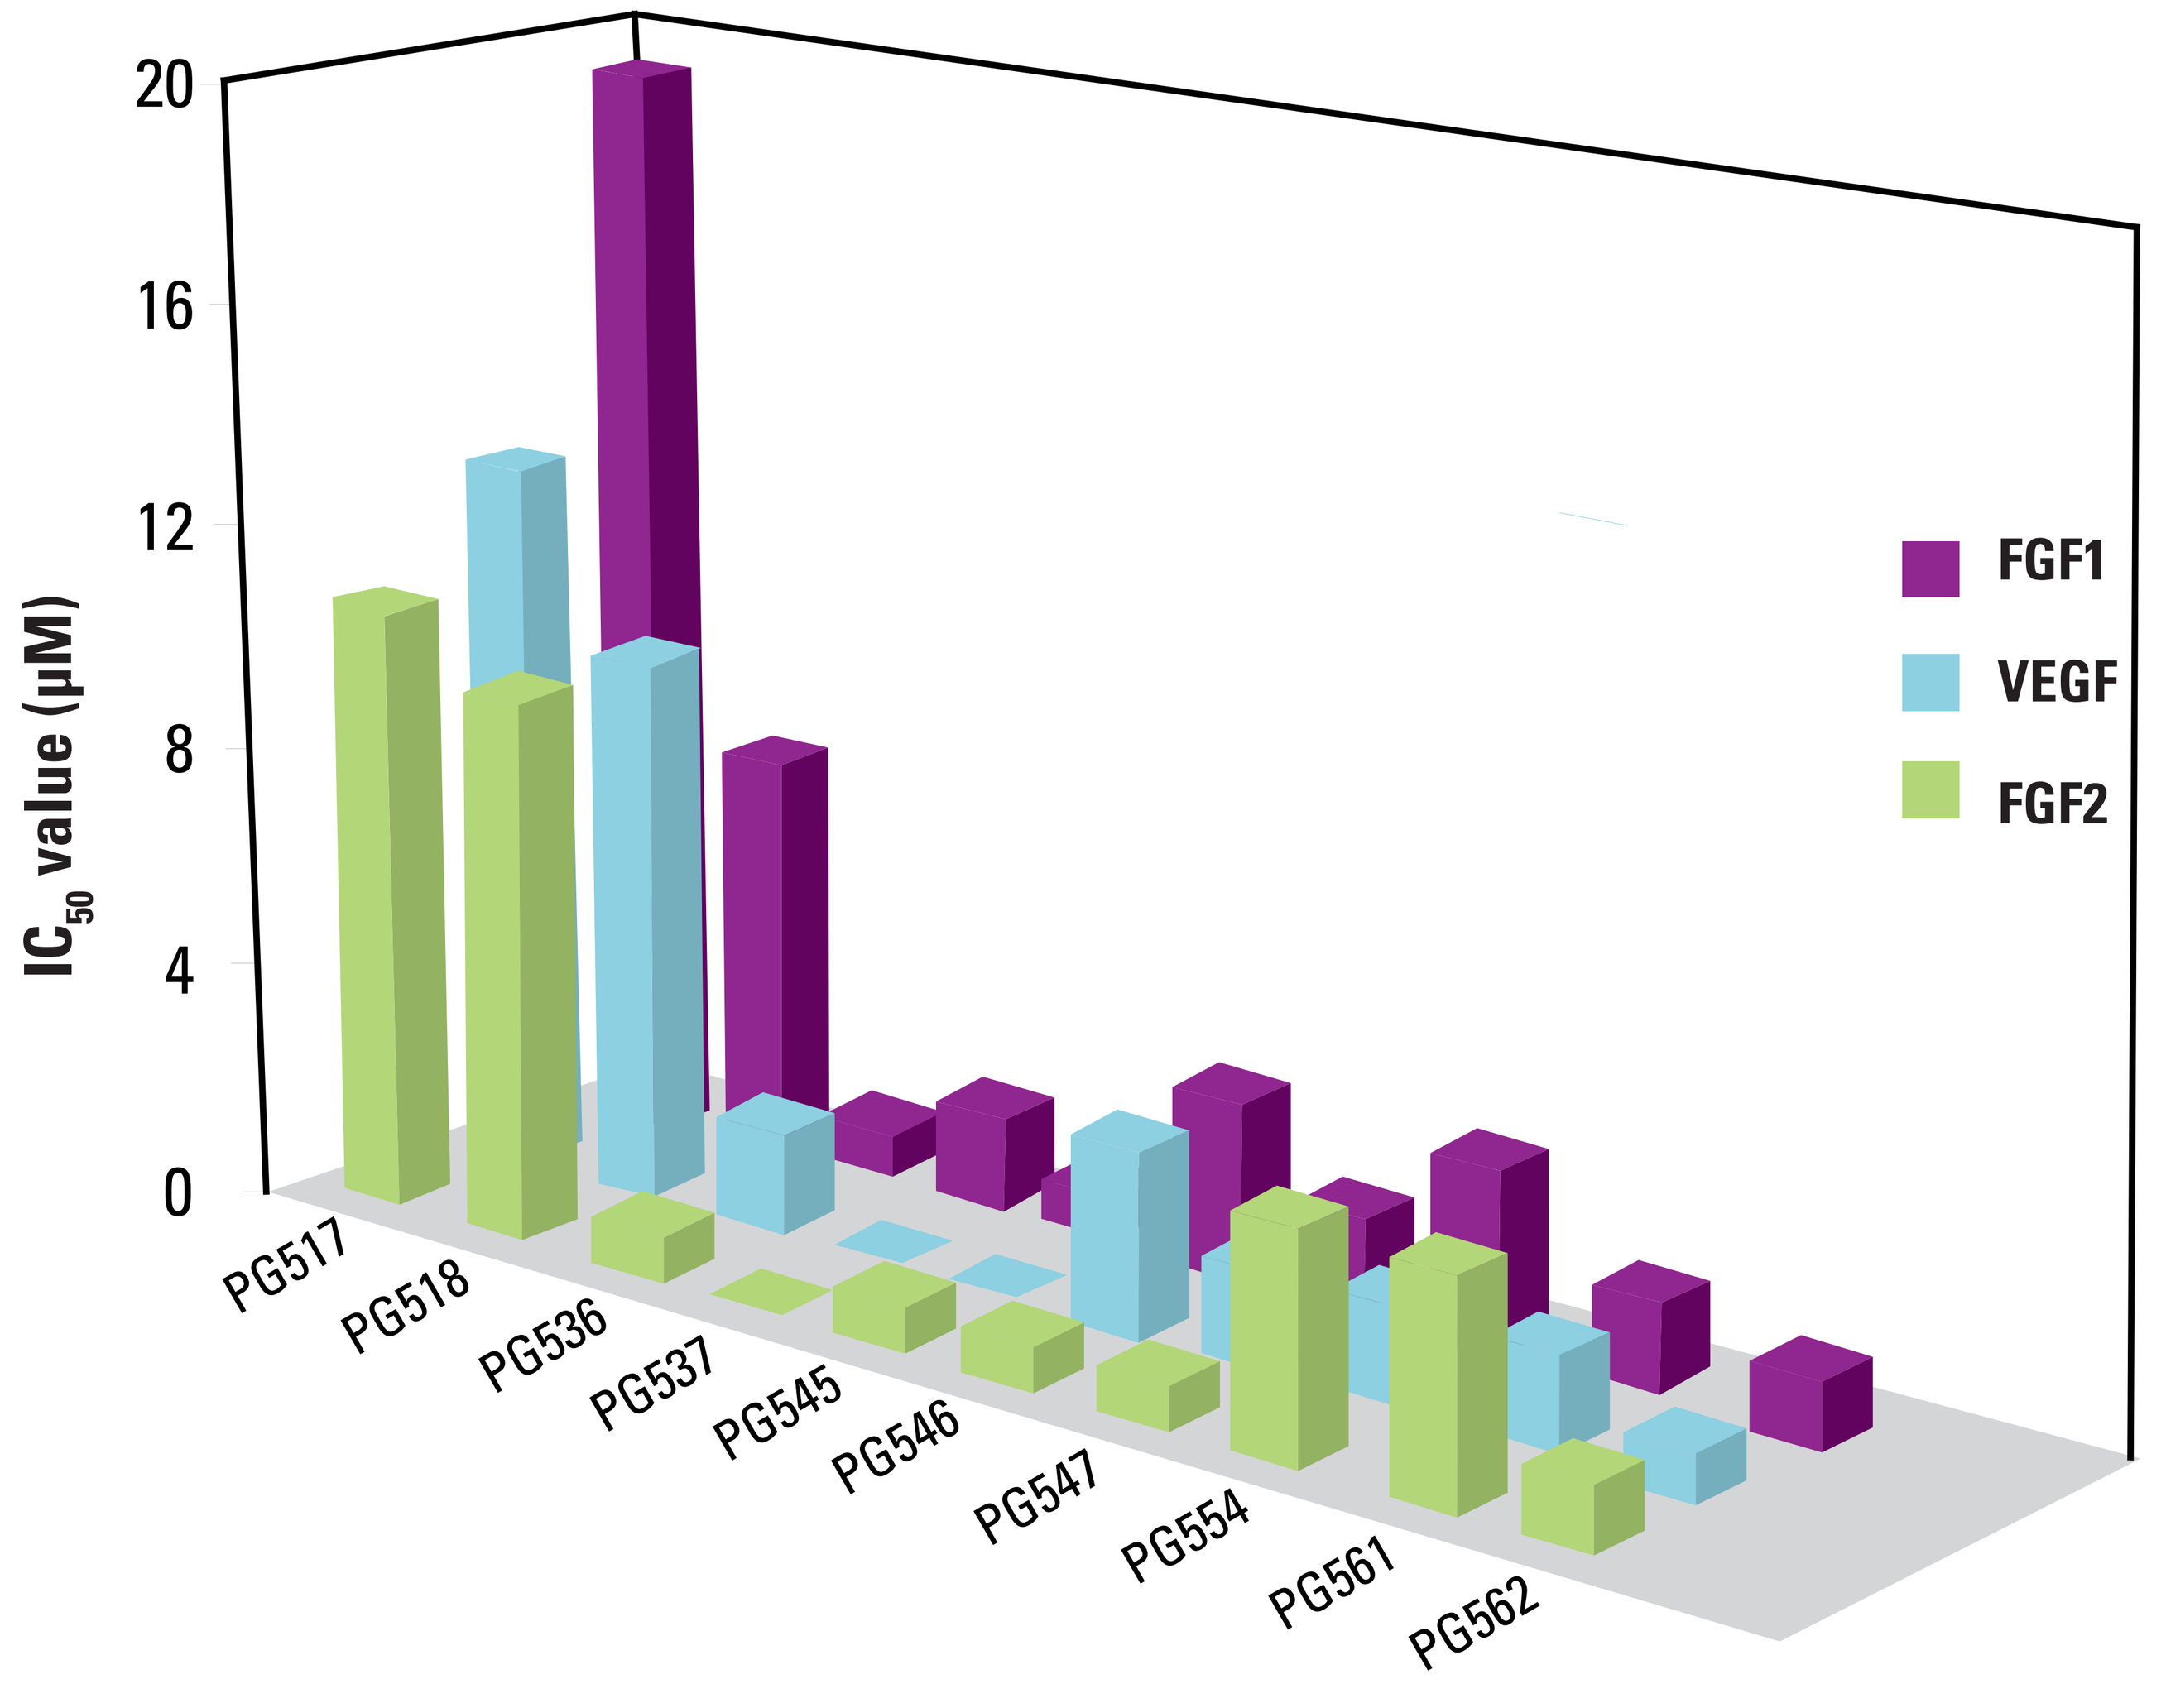
IC50 value (µM)
20
16
12
8
4
0
PG517
PG518
PG536
PG537
PG545
PG546
PG547
PG554
PG561
PG562
FGF1
VEGF
FGF2

Figure 1: IC_{50} values for selected PG500 series compounds in the HUVEC Proliferation Assay

Enhanced anti-proliferative activity was particularly evident in compounds from PG536 onwards. In many instances, similar values were obtained using endothelial cells of microvascular origin, dHMVEC (data not shown).

ENDOTHELIAL TUBE FORMATION

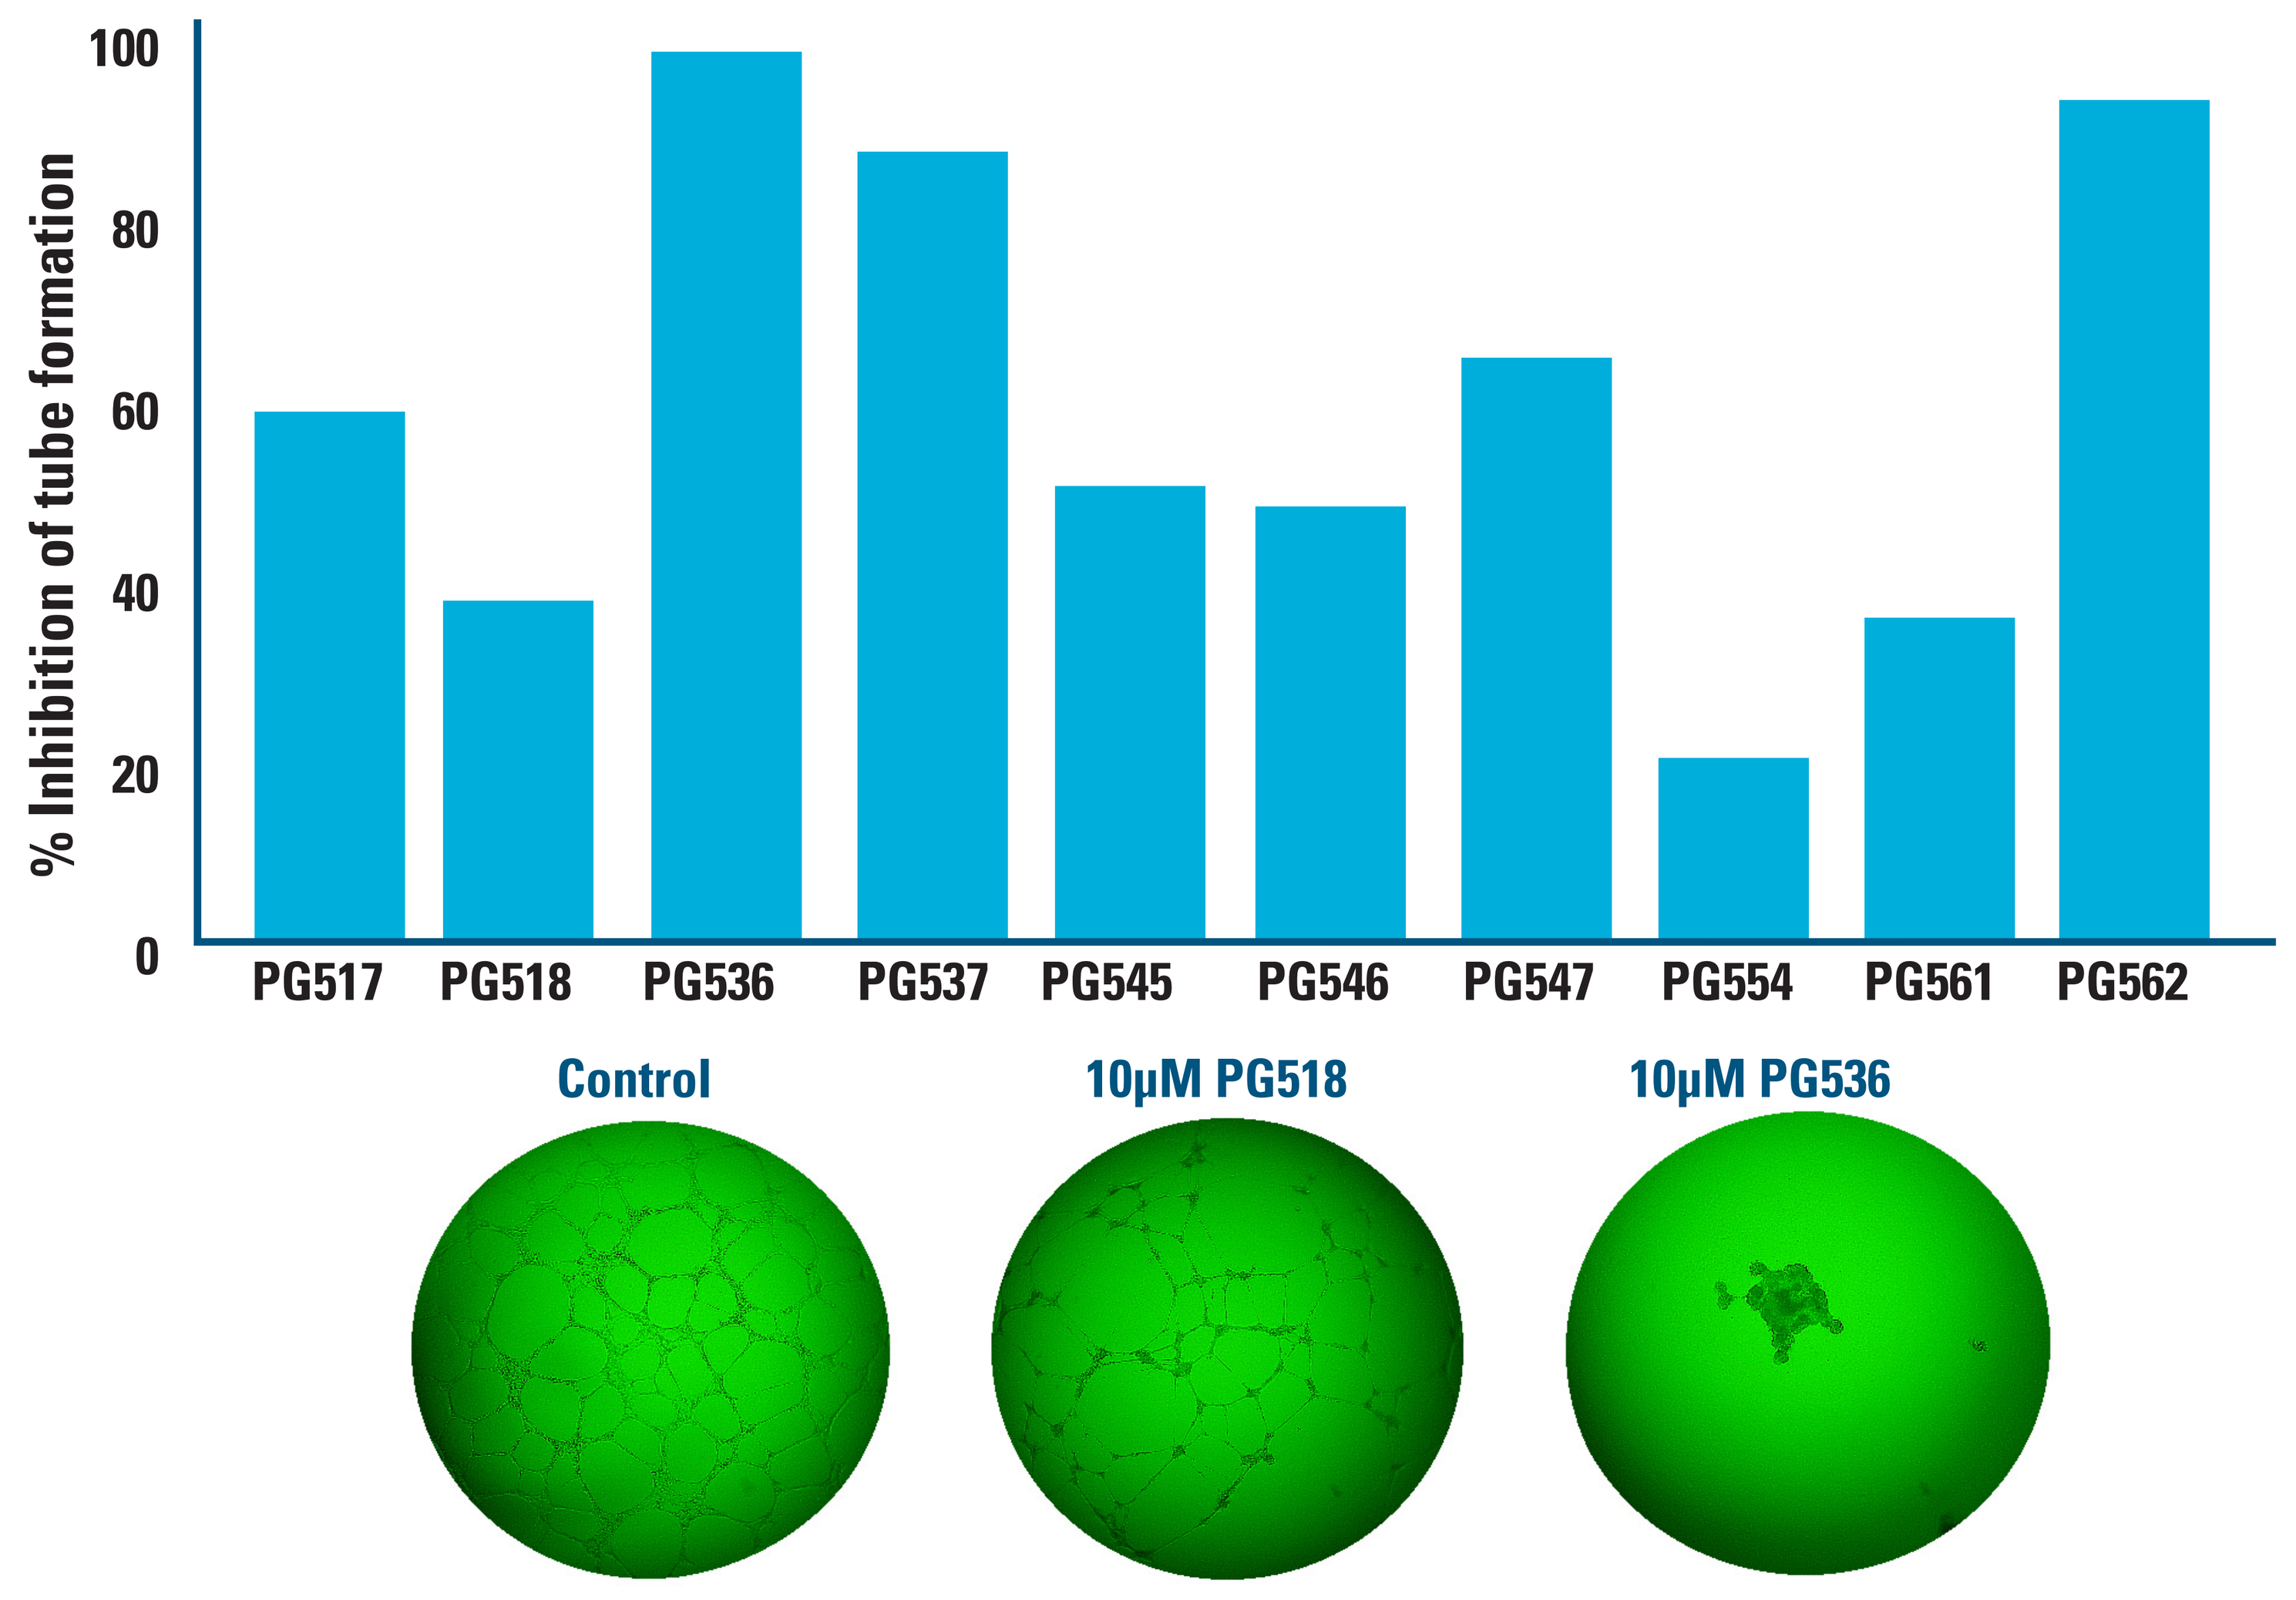
% Inhibition of tube formation
100
80
60
40
20
0
PG517
PG518
PG536
PG537
PG545
PG546
PG547
PG554
PG561
PG562
Control
10µM PG518
10µM PG536

Figure 2: Inhibition of HUVEC tube formation by selected PG500 series compounds at a concentration of 10µM.

Some representative images of control, PG518 (10µM) and PG536 (10µM) are also shown. Again, compounds tested in this assay using endothelial cells of microvascular origin (dHMVEC) exhibited similar inhibitory profiles (data not shown).

RAT AORTIC ASSAY FOR ANGIOGENESIS

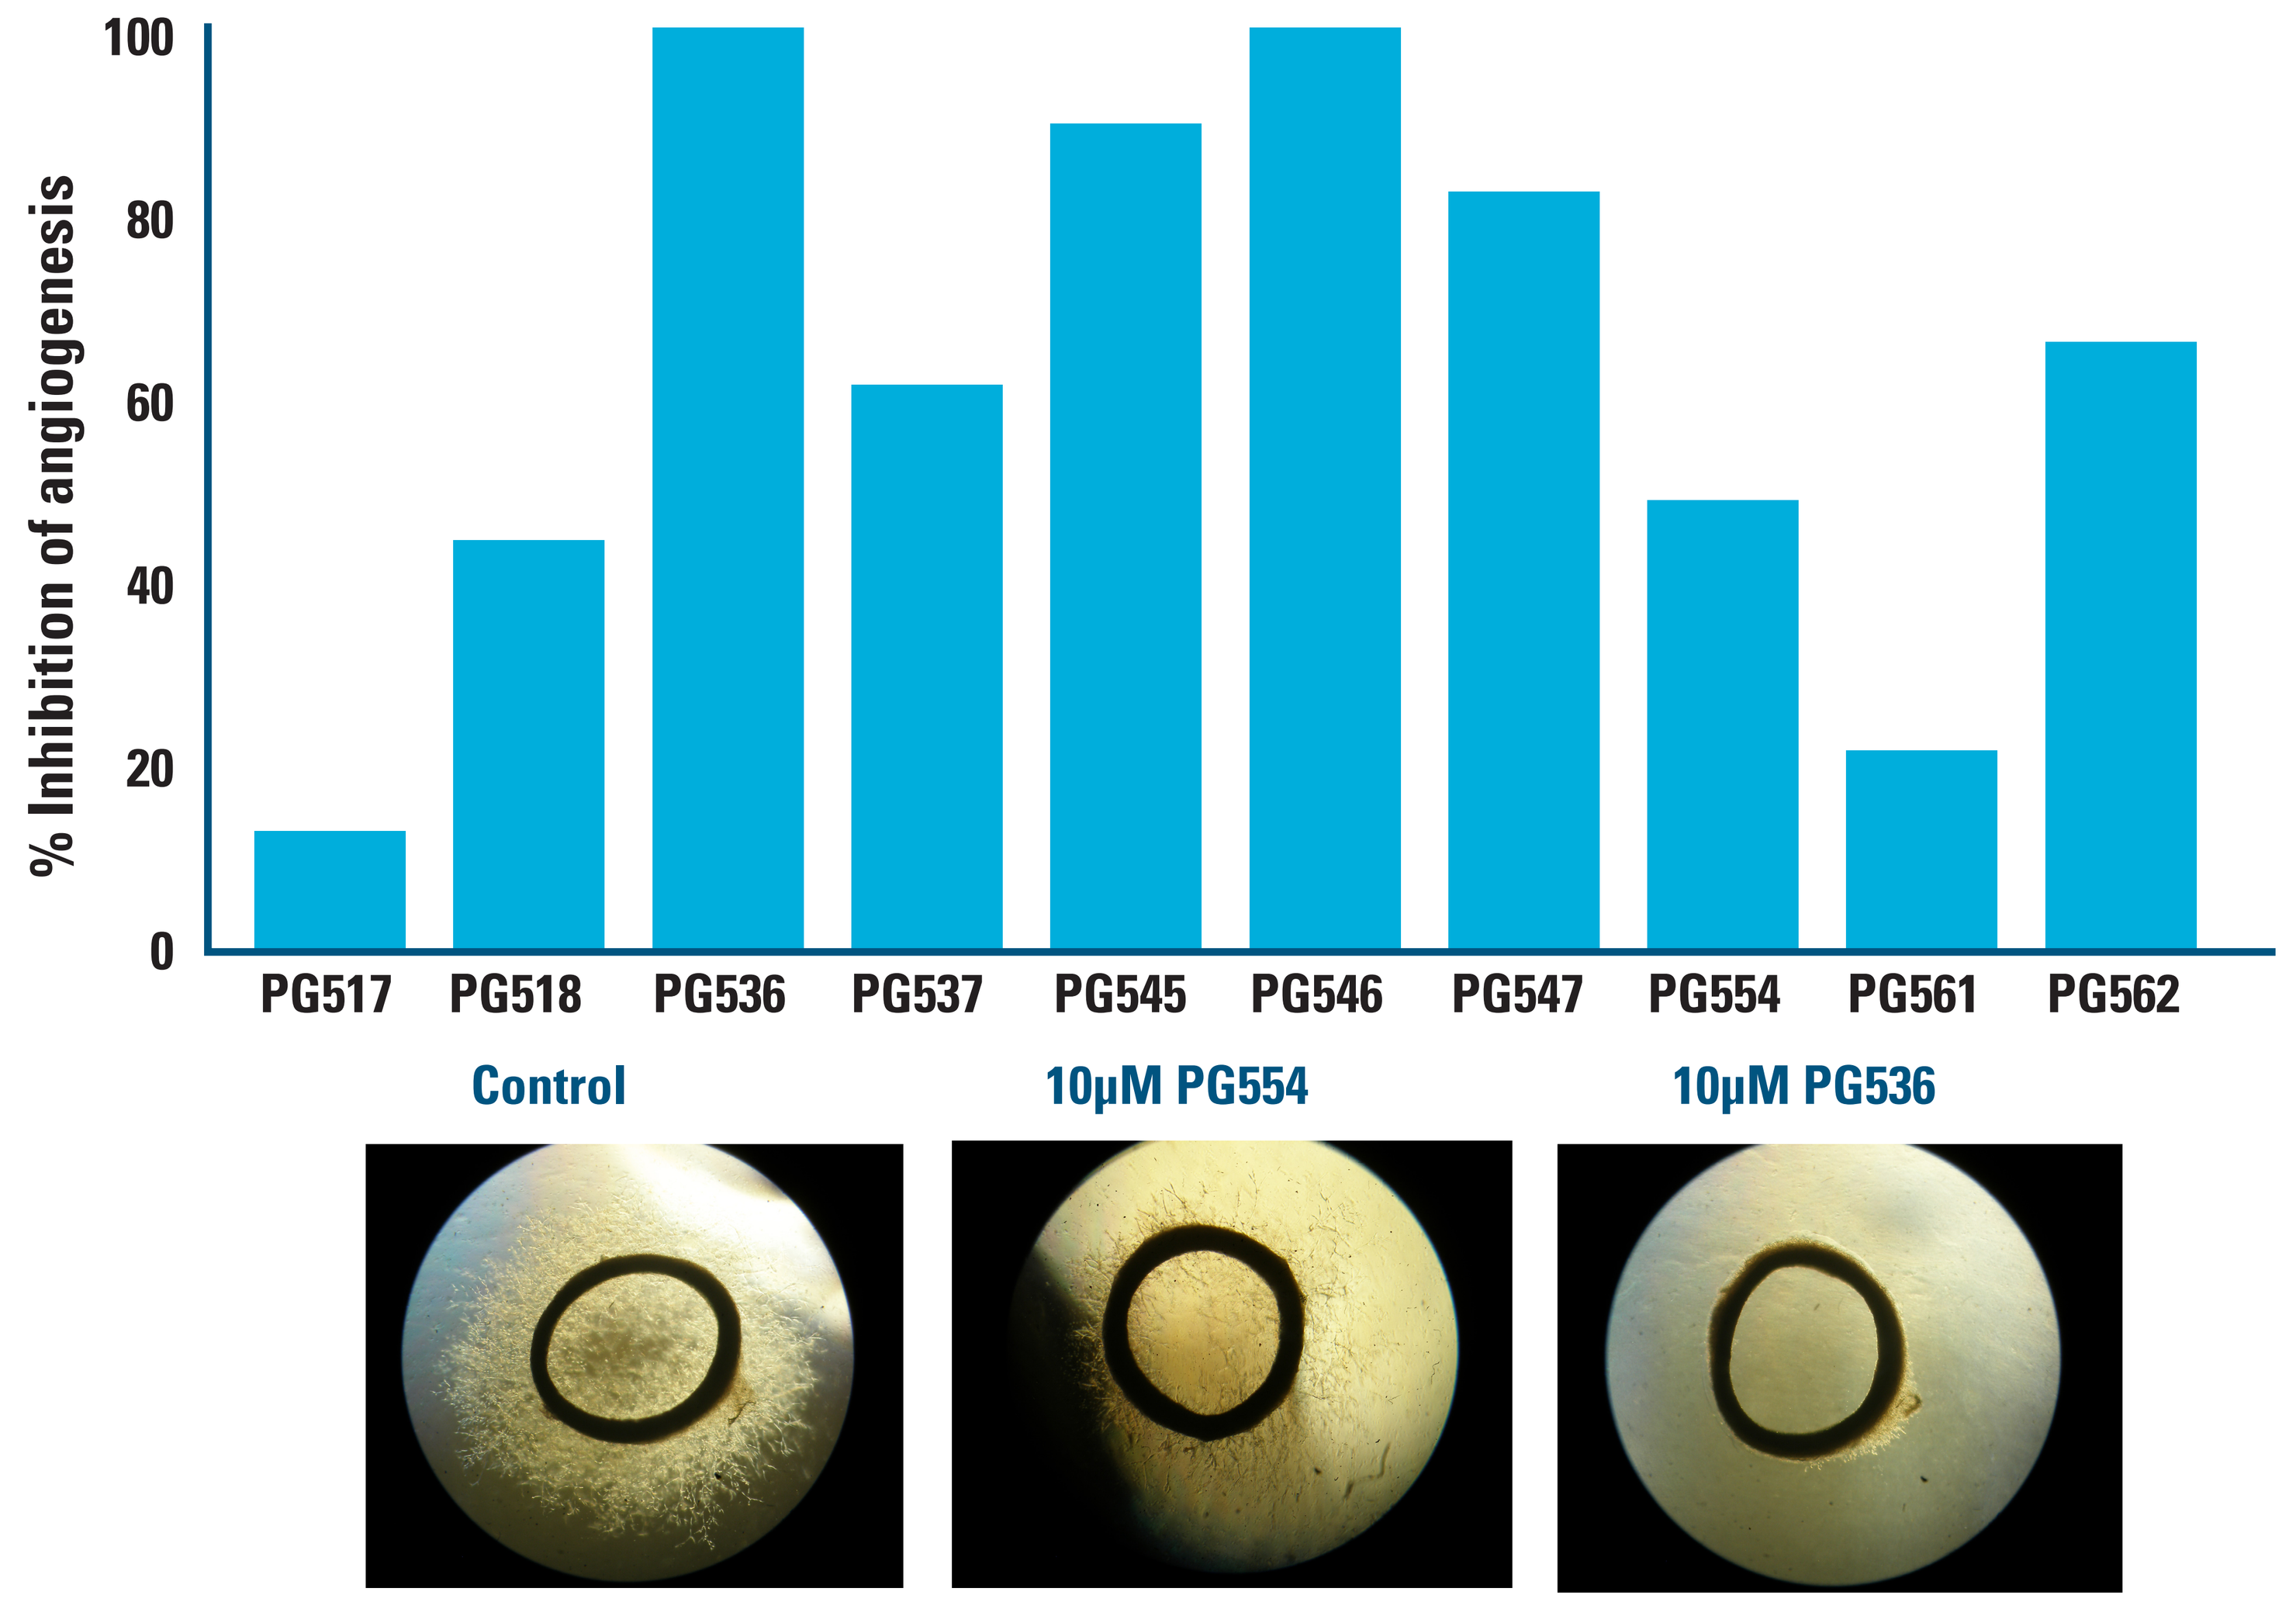
% Inhibition of angiogenesis
100
80
60
40
20
0
PG517
PG518
PG536
PG537
PG545
PG546
PG547
PG554
PG561
PG562
Control
10µM PG554
10µM PG536

Figure 3: Inhibition of angiogenesis by selected PG500 series compounds in the rat aortic assay

PG536, PG545 and PG546 were particularly potent at 10µM. PG536 was selected to examine the reversibility of the antiangiogenic effect by treating the aortic cultures with VEGF (10ng/ml) following a washout period. Recovery was evident after 72 h of culture (data not shown).

METHODS

- *In Vitro / Ex Vivo* Assays
 - HUVEC Proliferation
 - HUVEC Tube Formation (Matrigel™ Assay)
 - Rat Aortic Assay for Angiogenesis
- B16F1 Syngeneic Model for Melanoma
 - Antitumor activity tested in several PG500 series compounds
 - $5x10^5$ cells injected subcutaneously (sc) per mouse at day 0
 - Daily treatment (sc) for 12 days from day 3.
- HT29 Xenograft Model for Colorectal Cancer
 - Antitumor activity tested for selected PG500 series compounds
 - $2x10^6$ cells injected (sc) per mouse at day 0.
 - Daily treatment commenced once tumors reached $150mm^3$ for up to 3 weeks.

ANTI-TUMOR ACTIVITY AGAINST B16 MELANOMA

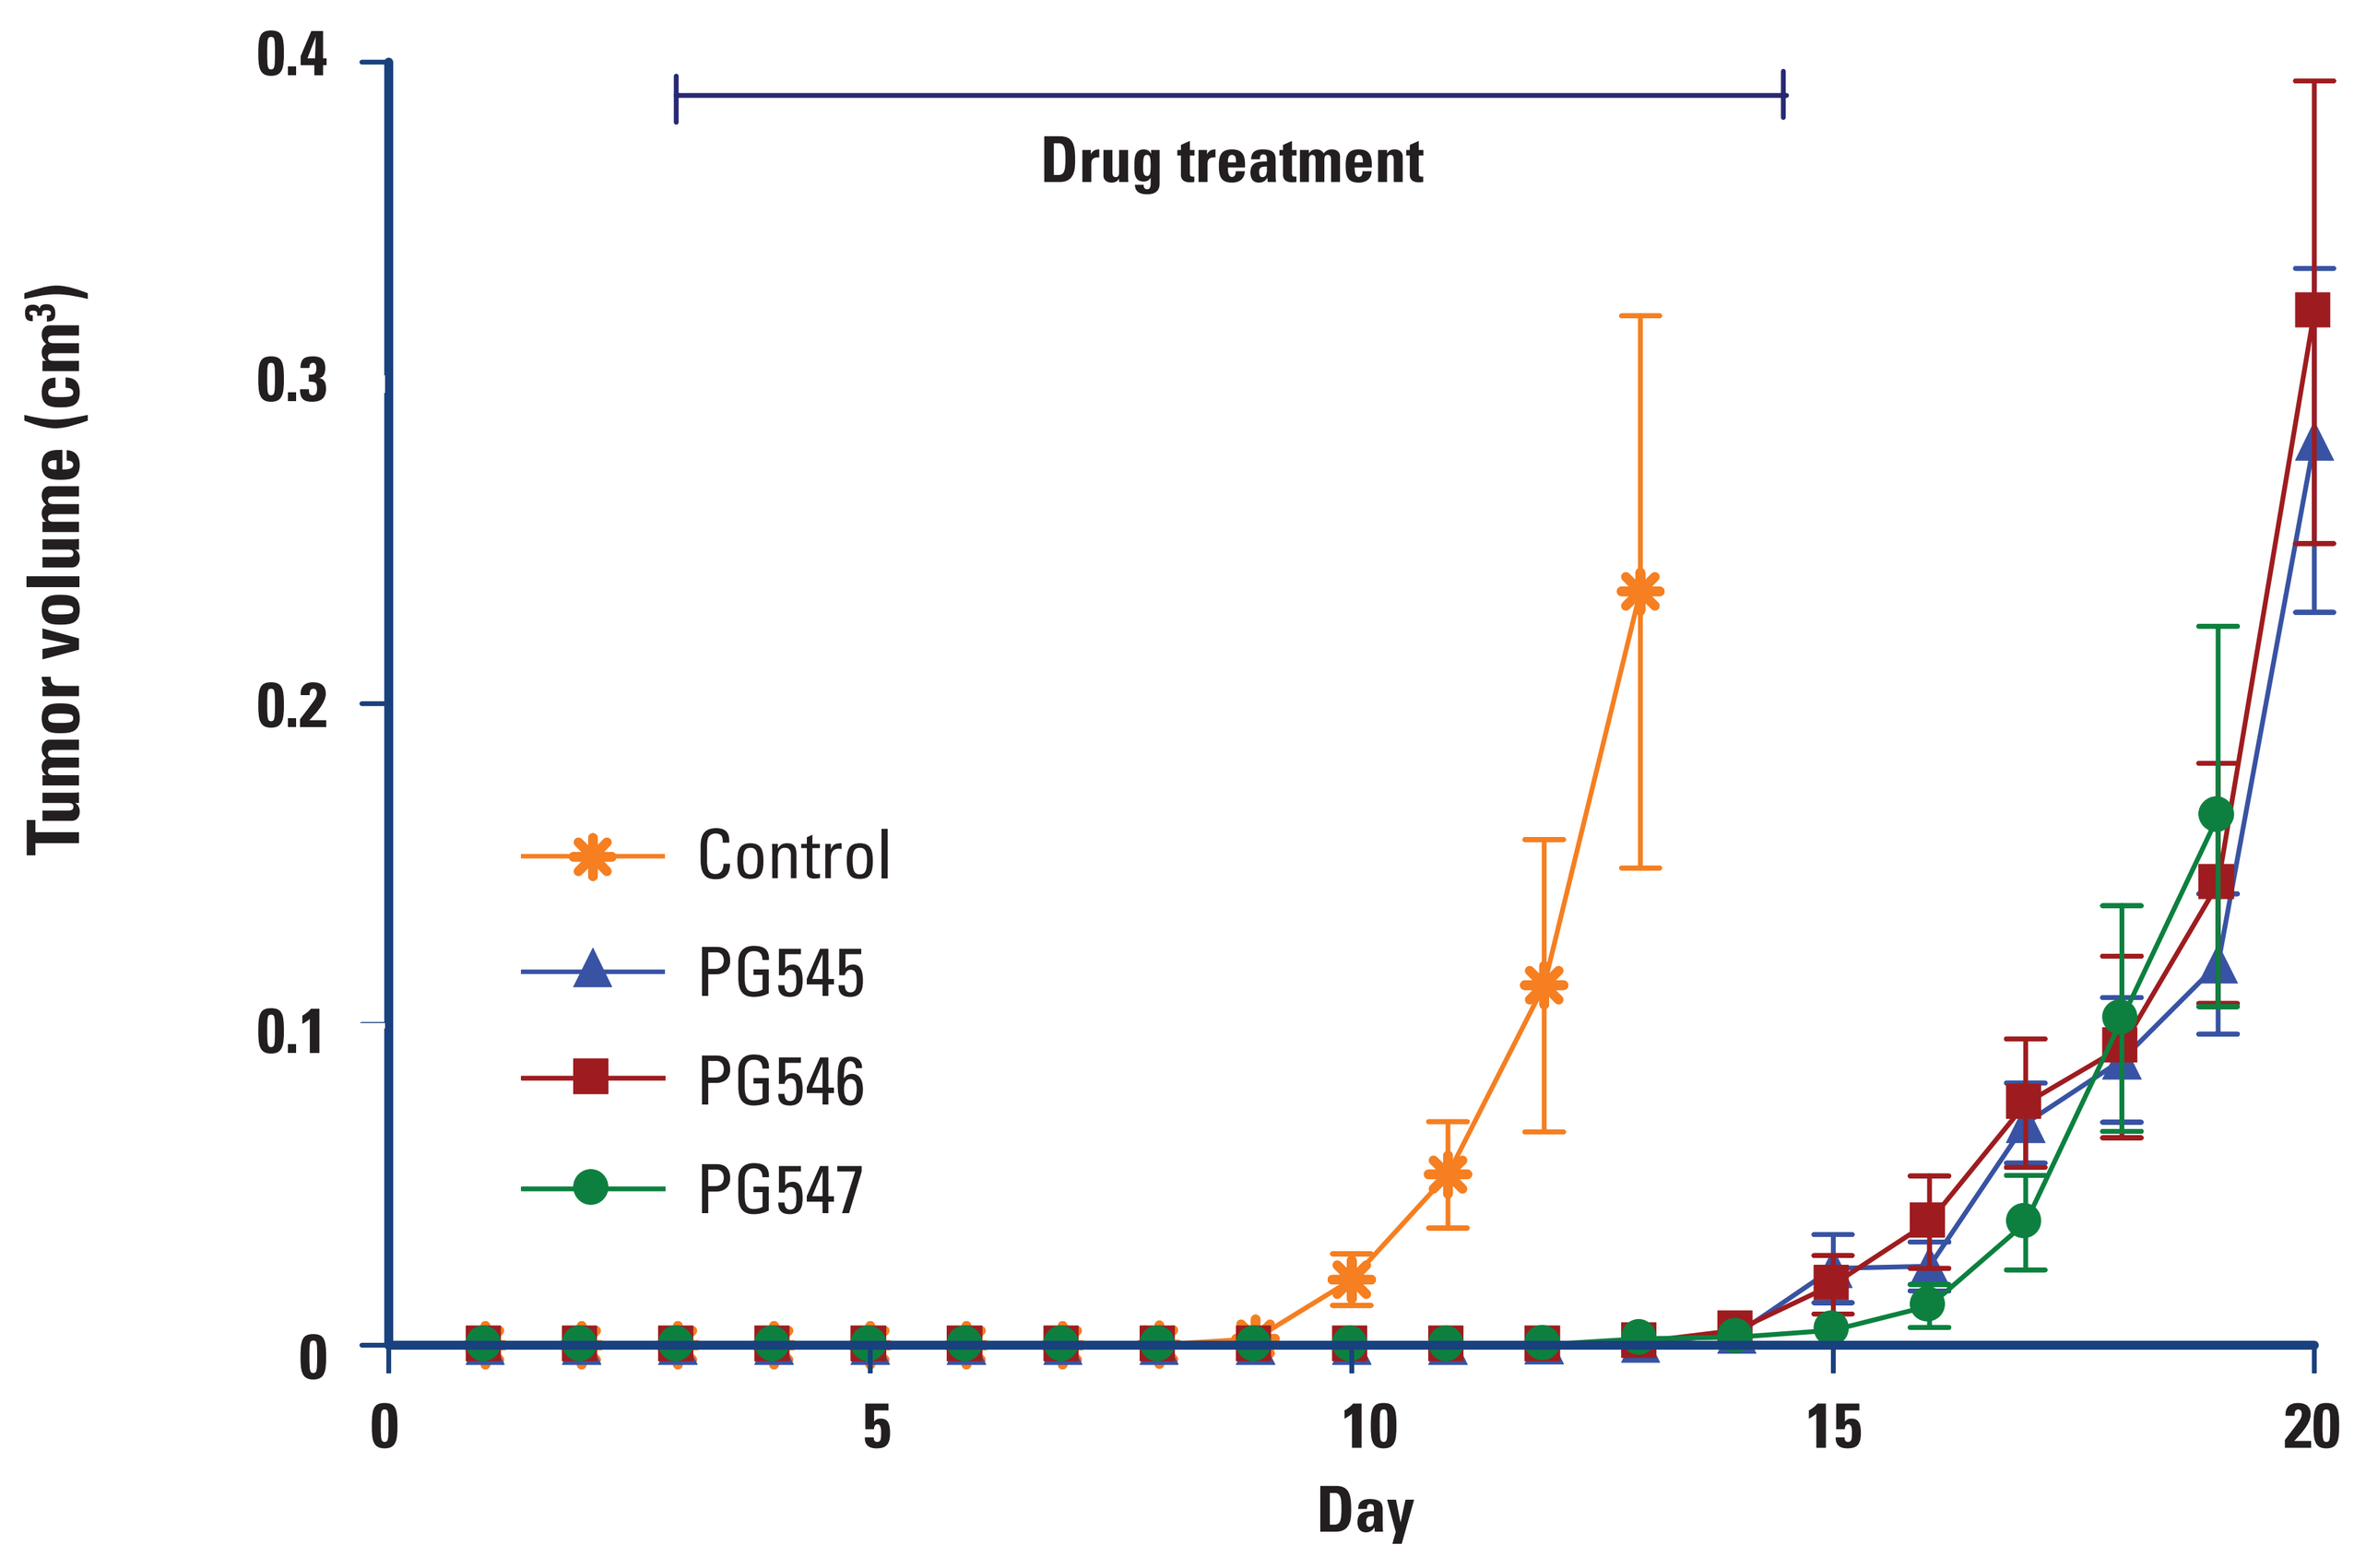
Tumor volume (cm3)
0.4
0.3
0.2
0.1
0
Drug treatment
Control
PG545
PG546
PG547
0
5
10
15
20
Day

Figure 4: PG500 series compounds significantly inhibit tumor progression in the B16F1 melanoma model.

Compounds were administered subcutaneously once daily for 12 days at 15 mg/kg/day, 3 days after tumor challenge at Day 0. During the period of administration, all three PG500 series compounds ensured the tumor was barely palpable. At the end of the treatment period similar tumor progression was observed in all groups.

ANTI-TUMOR ACTIVITY AGAINST HT29 COLORECTAL TUMOR

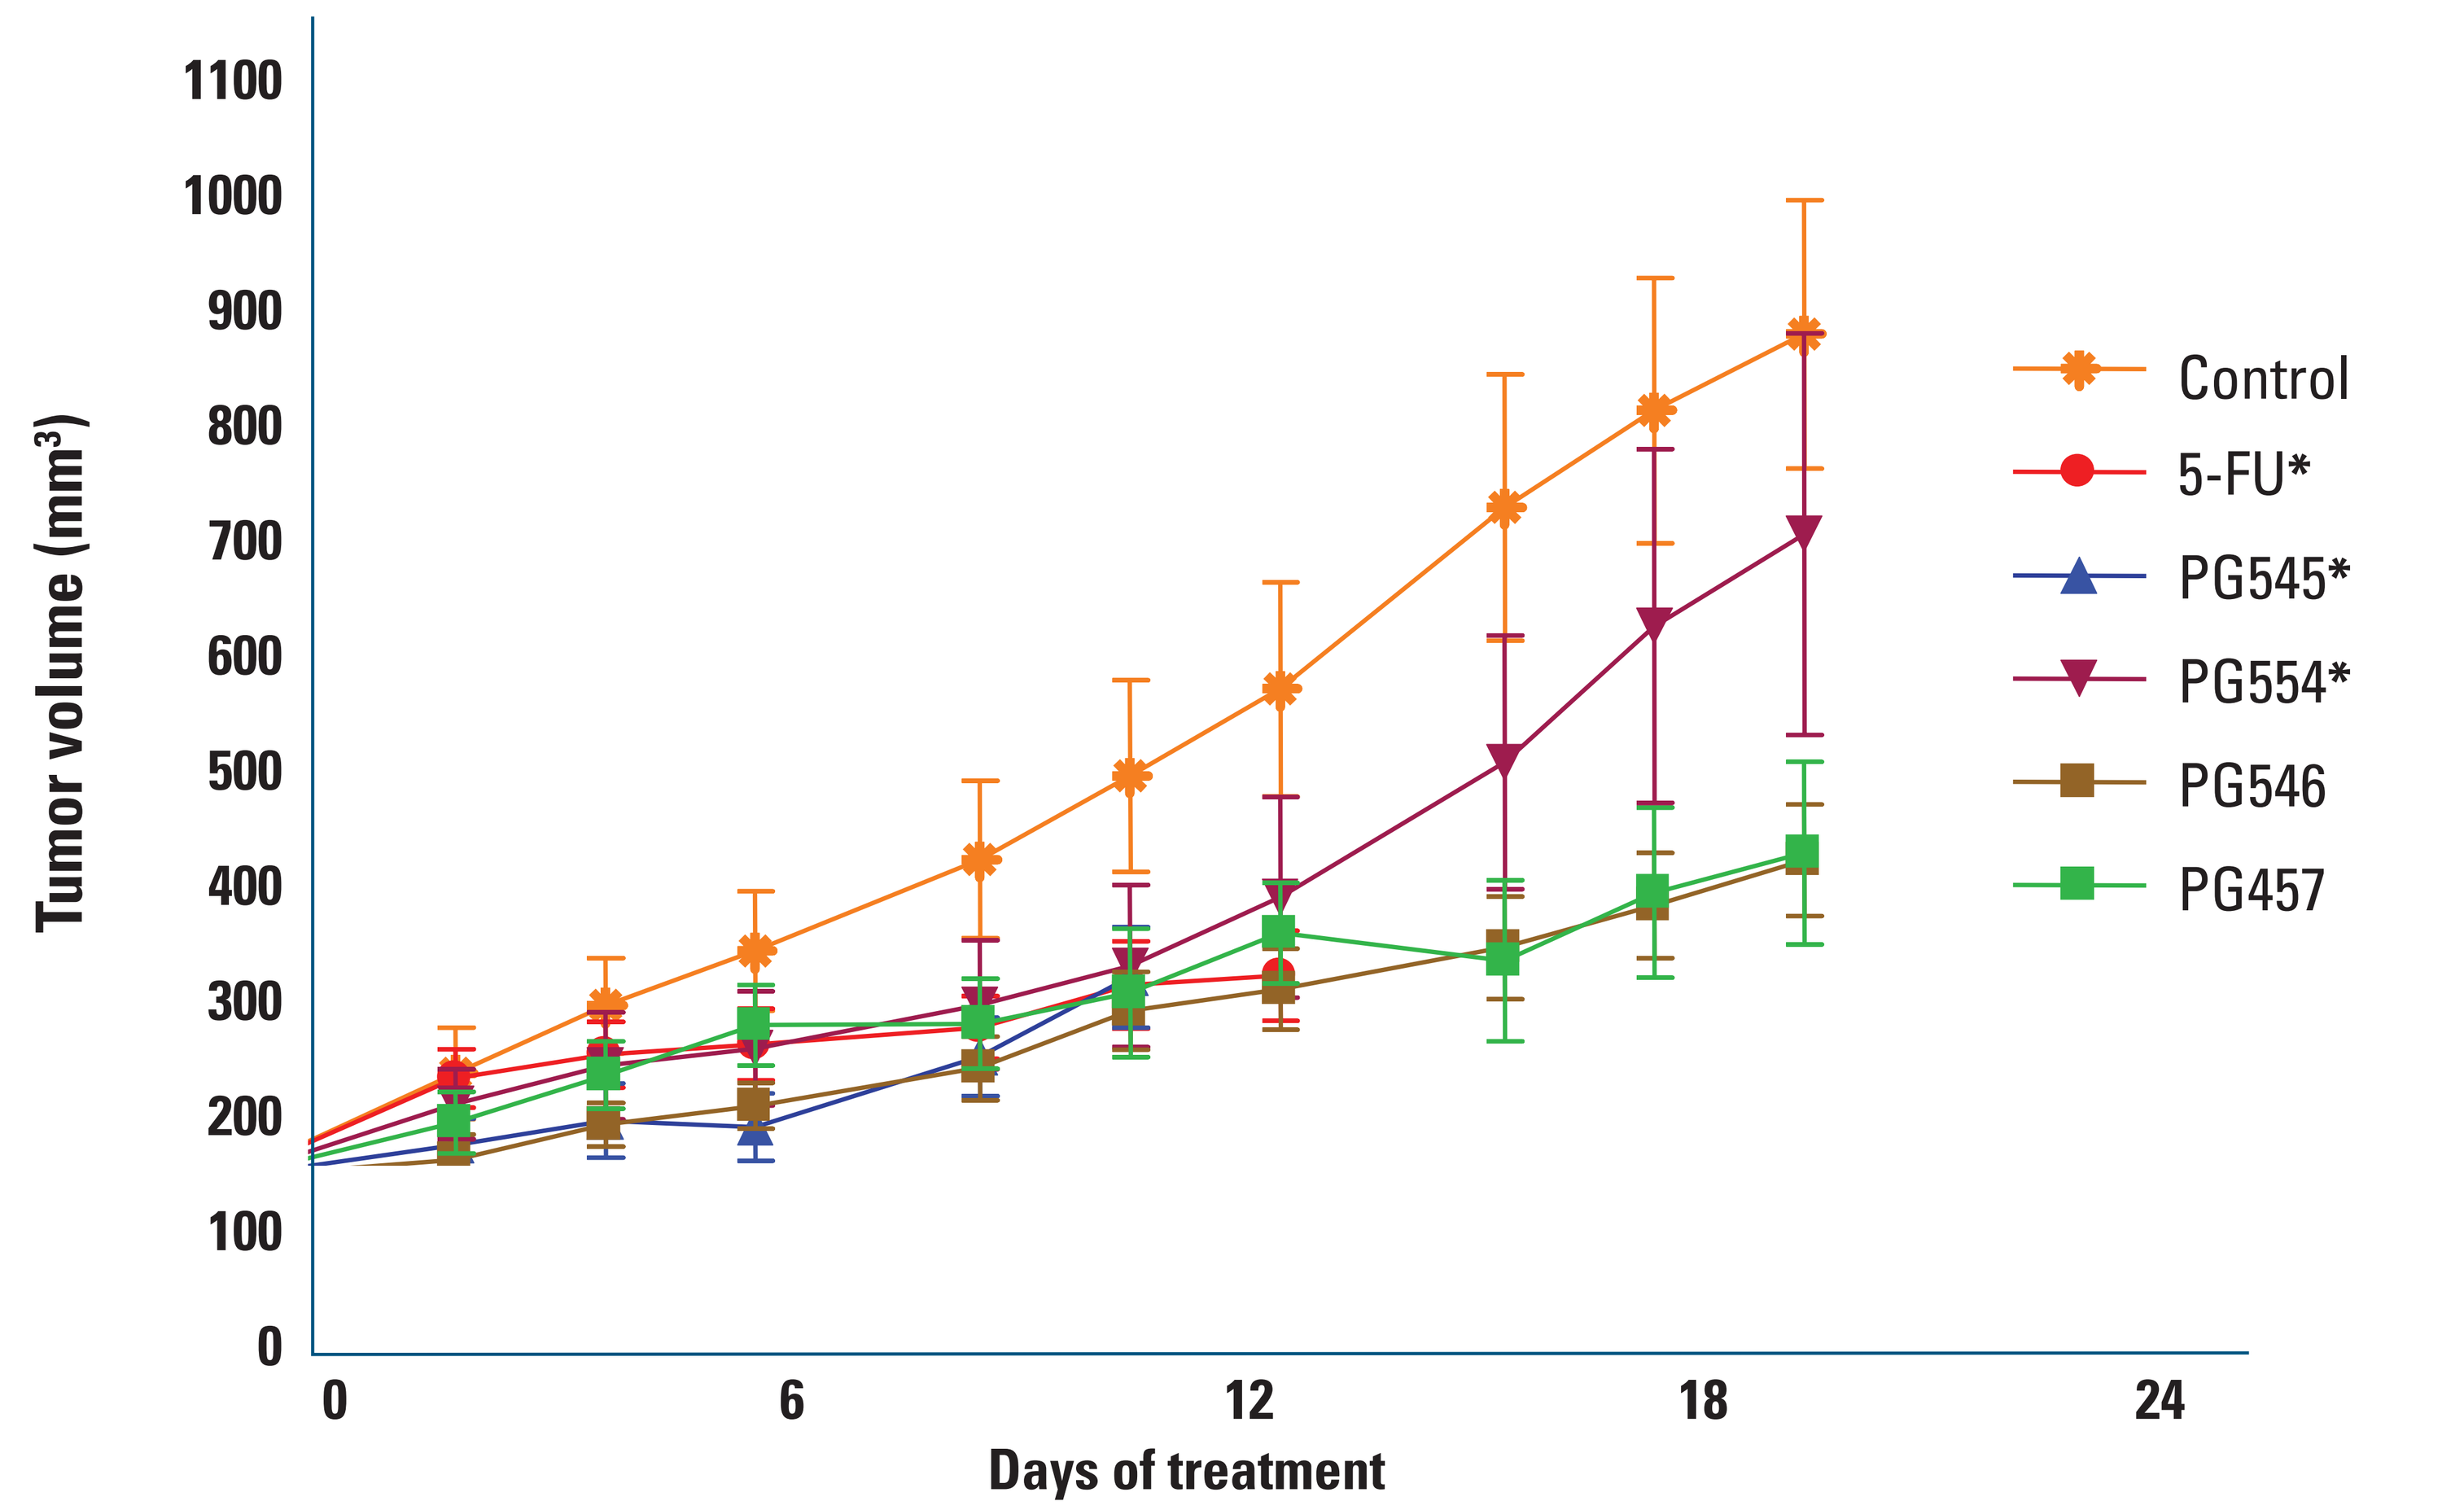
Tumor volume (mm3)
1100
1000
900
800
700
600
500
400
300
200
100
0
0
6
12
18
24
Days of treatment
Control
5-FU*
PG545*
PG554*
PG546
PG457

Figure 5: PG500 series compounds significantly inhibit tumor progression in the HT29 xenograft model.

Compounds were administered once daily at 10 mg/kg/day once the tumor reached a size of $150mm^3$. All PG500 series compounds exhibited an antitumor effect at least equivalent to 5-FU.

**Treatment with these compounds led to bodyweight loss and termination of treatment between days 11-14.*

SUMMARY

- Several PG500 series compounds exhibited potent biological activity in the following studies:
 - Inhibition of growth factor-induced proliferation
 - Inhibition of tube formation
 - Inhibition of angiogenesis (in rat aortic assay)
- Potent anti-tumor activity observed in two models:
 - B16F1 melanoma model
 - HT29 colorectal xenograft model
- The data strongly supports continued preclinical development of this series of HS mimetics as anti-cancer agents

ACKNOWLEDGEMENTS

Progen would like to thank the following contributors of this work.

- Prof. Tom Gonda (Diamantina Institute for Cancer, Immunology and Metabolic Medicine, University of Qld, Australia)
- Dr. Ralf Brandt (vivoPharm Pty Ltd, Adelaide, Australia)